Guselkumab Binding to CD64⁺ IL-23–producing Myeloid Cells Enhances Potency for Neutralizing IL-23 Signaling

P2581

Jessica R. Allegretti,[1,*] Raja Atreya,[2] Maria T. Abreu,[3] Amy Hart,[4] He (Hurley) Li,[4] Tom C. Freeman,[4] Eilyn Lacy,[4] Matthew DuPrie,[4] Phuc Bao,[5] Tina Smets,[6] Joshua Wertheimer,[4] Indra Sarabia,[5] Kristen Kohler,[5] Anne Fourie,[5] Kacey Sachen[5]

[1]Division of Gastroenterology, Hepatology and Endoscopy, Department of Medicine, Brigham and Women's Hospital, Harvard Medical School, Boston, MA, USA; [2]Department of Medicine 1, Erlangen University Hospital, Friedrich-Alexander-Universität Erlangen-Nürnberg, Erlangen, Germany; [3]Leonard M. Miller School of Medicine, University of Miami, Miami, FL, USA; [4]Janssen Research & Development, LLC, Spring House, PA, USA; [5]Janssen Research & Development, LLC, San Diego, CA, USA; [6]Janssen Research & Development, Beerse, Belgium.

*Presenting author.

This material is distributed for scientific purposes on Janssen Science, and is not for promotional use

Background

- Interleukin (IL)-23 is implicated in the pathogenesis of inflammatory bowel disease (IBD), and myeloid cells that express Fcγ receptor (FcγR) I (CD64) have been identified as the primary cellular source of IL-23 in inflamed IBD gut tissue[1,2]
- Guselkumab (GUS), risankizumab (RZB), and mirikizumab (MIRI) are monoclonal antibodies (mAbs) specifically directed against the IL-23p19 subunit (illustration to the right)
 - GUS is a fully human immunoglobulin (Ig) G1 mAb with a native Fc region[3]
 - RZB is a humanized IgG1 mAb with leucine to alanine substitutions at positions 234 and 235 (LALA) in the Fc region that diminish binding to FcγRs[4]
 - MIRI is a humanized IgG4 mAb with a phenylalanine to alanine substitution at position 234 and a leucine to alanine substitution at position 235 (FALA) in the Fc region that diminish binding to FcγRs[5,6]
- GUS, RZB, and MIRI have shown efficacy in the treatment of IBD[3,4,7-9]; however, potential differences may exist in the therapeutic profiles of GUS, RZB, and MIRI

	GUS	RZB	MIRI
Antibody	Fully human	Humanized	Humanized
Isotype	IgG1	IgG1	IgG4
Antigen	IL-23p19	IL-23p19	IL-23p19
Fc domain	Native/wild type	Mutated (LALA)	Mutated (FALA)

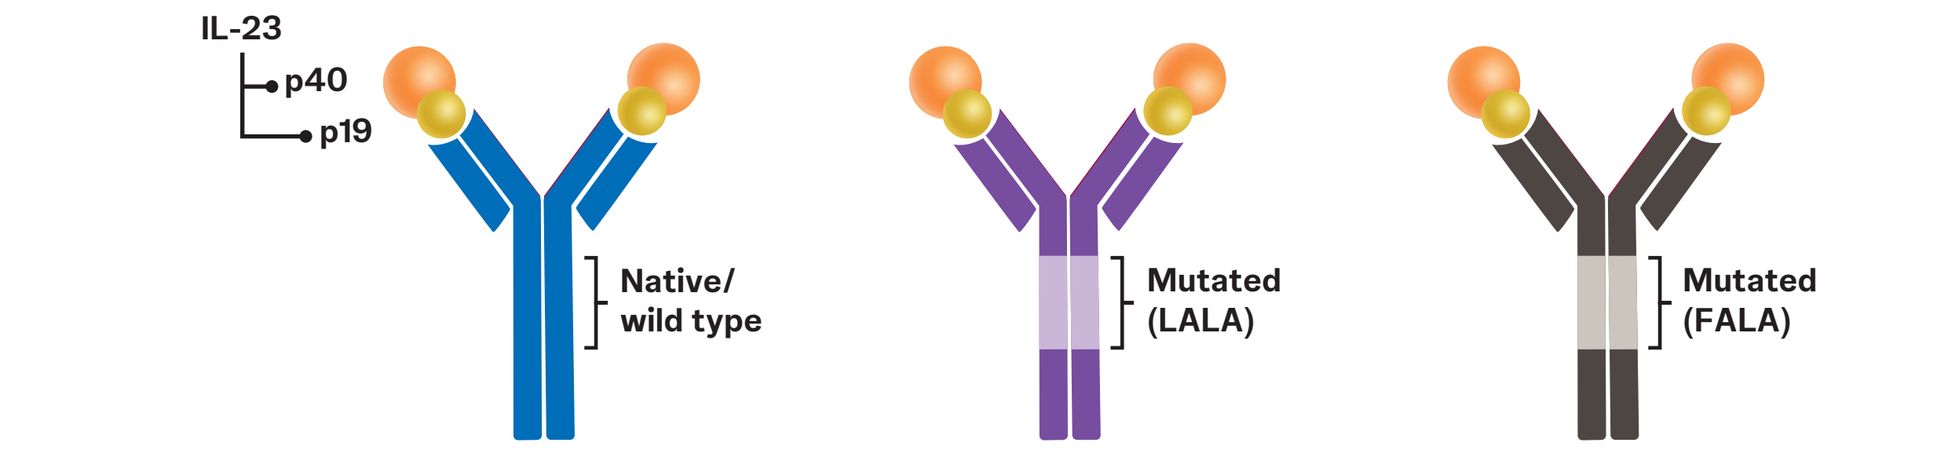
IL-23
p40
p19
Native/
wild type
Mutated
(LALA)
Mutated
(FALA)

Objective

Here, we evaluated CD64 and IL-23 expression in IBD patient gut biopsies; the binding of GUS, RZB, and MIRI to CD64; and the functional consequences of CD64 binding by IL-23p19 subunit mAbs in in vitro assays

Methods

Bulk Transcriptomic Analyses

- IBD RNA sequencing (RNAseq) data were obtained from GSE193677[10]
- Read counts from rectal biopsies of patients with Crohn's disease (CD), patients with ulcerative colitis (UC), and non-IBD controls were analyzed
- Data were converted to $\log_2$ counts per million (CPM), with the edgeR package (Bioconductor) used for plotting and statistical analysis[11]

Single-cell Transcriptomic Analyses

- Single-cell transcriptome data were obtained from previous literature that investigated CD (GSE134809)[12]
- SCANPY, a large-scale, single-cell gene-expression data-analysis tool, was used for single-cell analyses and visualization[13]
- Raw counts for each gene were normalized to the total counts per cell followed by log transformation to represent gene-expression levels

mAb Binding Affinity for IL-23: Kinetic Exclusion Assay (KinExA)

- Serial dilutions of recombinant human IL-23 (rhIL-23) were prepared in the presence of a constant concentration of mAbs. Assays were performed using 4 different mAb concentrations
- Titrations of mAb–IL-23 complexes were incubated at room temperature (~22°C) to reach binding equilibrium
- A KinExA 3200 or 4000 instrument measured the amount of free mAb in the reaction. Data were fit with a 1:1 binding model using KinExA Pro software

mAb Binding to CD64: Homogeneous Time-resolved Fluorescence (HTRF) Assay

- The binding of GUS, RZB, and MIRI to CD64 was assessed using an HTRF assay (PerkinElmer, Cisbio) with cells expressing recombinant CD64
- GUS, RZB, MIRI, and human IgG1 isotype control (hIgG1 IC) were assessed for their ability to bind to CD64 by competing with a control D2-labeled IgG antibody that induces an HTRF signal when bound to CD64-expressing cells

mAb Potency for Inhibition of IL-23 Signaling in Human Peripheral Blood Mononuclear Cells (PBMCs)

- STAT3 is a signal transducer and activator of transcription involved in IL-23 receptor proximal signaling
- Human PBMCs were expanded with a CD3-activating antibody in medium that was supplemented with IL-1β for 4 days
- PBMCs were stimulated for 30 minutes with 5 ng/mL rhIL-23 heterodimer in the presence or absence of GUS, RZB, MIRI, or hIgG1 IC
- PBMCs were lysed for measurement of phosphorylated STAT3 (pSTAT3) using a Meso Scale Discovery assay

IL-23 Receptor Bioassay

- IL-23 bioassay cells (Promega) are engineered to express the IL-23 receptor and a luciferase reporter gene under a STAT3-inducible promoter, connecting IL-23 receptor signaling to a luminescent signal
- THP-1 is a human monocyte cell line with constitutive expression of CD64. Like primary human monocytes, CD64 can be upregulated following priming with interferon γ (IFNγ), and IL-23 is produced following stimulation with R848
 - Conditioned medium containing native IL-23 was obtained from THP-1 cells that were primed with IFNγ and then stimulated with R848 for 18 hours
- To test the potency of IL-23p19 mAbs against endogenously secreted IL-23 in conditioned medium, GUS, RZB, MIRI, and hIgG1 IC were pre-incubated with the conditioned medium for 1 hour and then added to IL-23 bioassay cells. At 5 hours, IL-23 signaling was assessed by the addition of Bio-Glo™ luciferase substrate (Promega) and the measurement of luminescent signal (relative light units [RLU])
- To test the potency of IL-23p19 mAbs against endogenous IL-23 that is secreted locally by CD64 cells, a co-culture of IL-23 bioassay cells and IFNγ-primed THP-1 cells was stimulated with R848 for 16 hours in the presence or absence of GUS, RZB, MIRI, or hIgG1 IC, and luminescent signal was measured as previously described

Key Takeaways

- Our transcriptomic analysis supports previous observations that CD64⁺ myeloid cells are a key source of IL-23 production in inflamed IBD gut tissue
- GUS binds to CD64, while RZB and MIRI do not bind to CD64 by virtue of their mutated Fc regions
- GUS, RZB, and MIRI bind the IL-23p19 subunit and neutralize IL-23 with similar high potency when IL-23 is exogenously added to IL-23–responsive cells
- GUS displays enhanced potency for the neutralization of IL-23 signaling in a co-culture of IL-23–producing CD64⁺ myeloid cells with IL-23–responsive cells compared to RZB and MIRI
- These in vitro data support a hypothesis for optimal localization of GUS in inflamed tissues where CD64⁺ IL-23–producing myeloid cells are increased and in proximity to IL-23–responsive lymphoid cells, which may contribute to the maintenance of clinical response and therapeutic differences within the IL-23p19 subunit inhibitor class
 - Additional clinical studies will be required to determine how these in vitro observations translate to clinically meaningful differences for patients with IL-23–mediated diseases

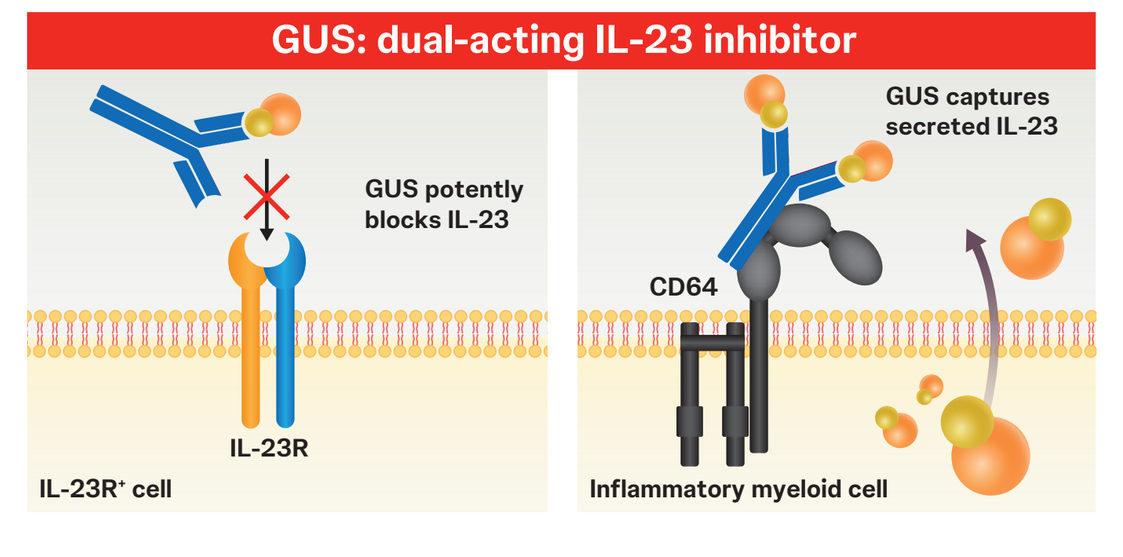
GUS: dual-acting IL-23 inhibitor
GUS potently blocks IL-23
GUS captures secreted IL-23
CD64
IL-23R
IL-23R⁺ cell
Inflammatory myeloid cell

Results

Expression of *FCGR1A* (CD64), *IL23A* (IL-23p19), and *IL12B* (IL-23p40) was significantly increased in inflamed versus non-inflamed IBD gut biopsies (Figure 1), and *IL23A*-expressing myeloid cells co-express *FCGR1A* and are increased in involved tissue (Figure 2)

Figure 1. *FCGR1A* (CD64), *IL23A* (IL-23p19), and *IL12B* (IL-23p40) expression from bulk transcriptomic datasets of rectal biopsies from patients with CD, patients with UC, and non-IBD controls

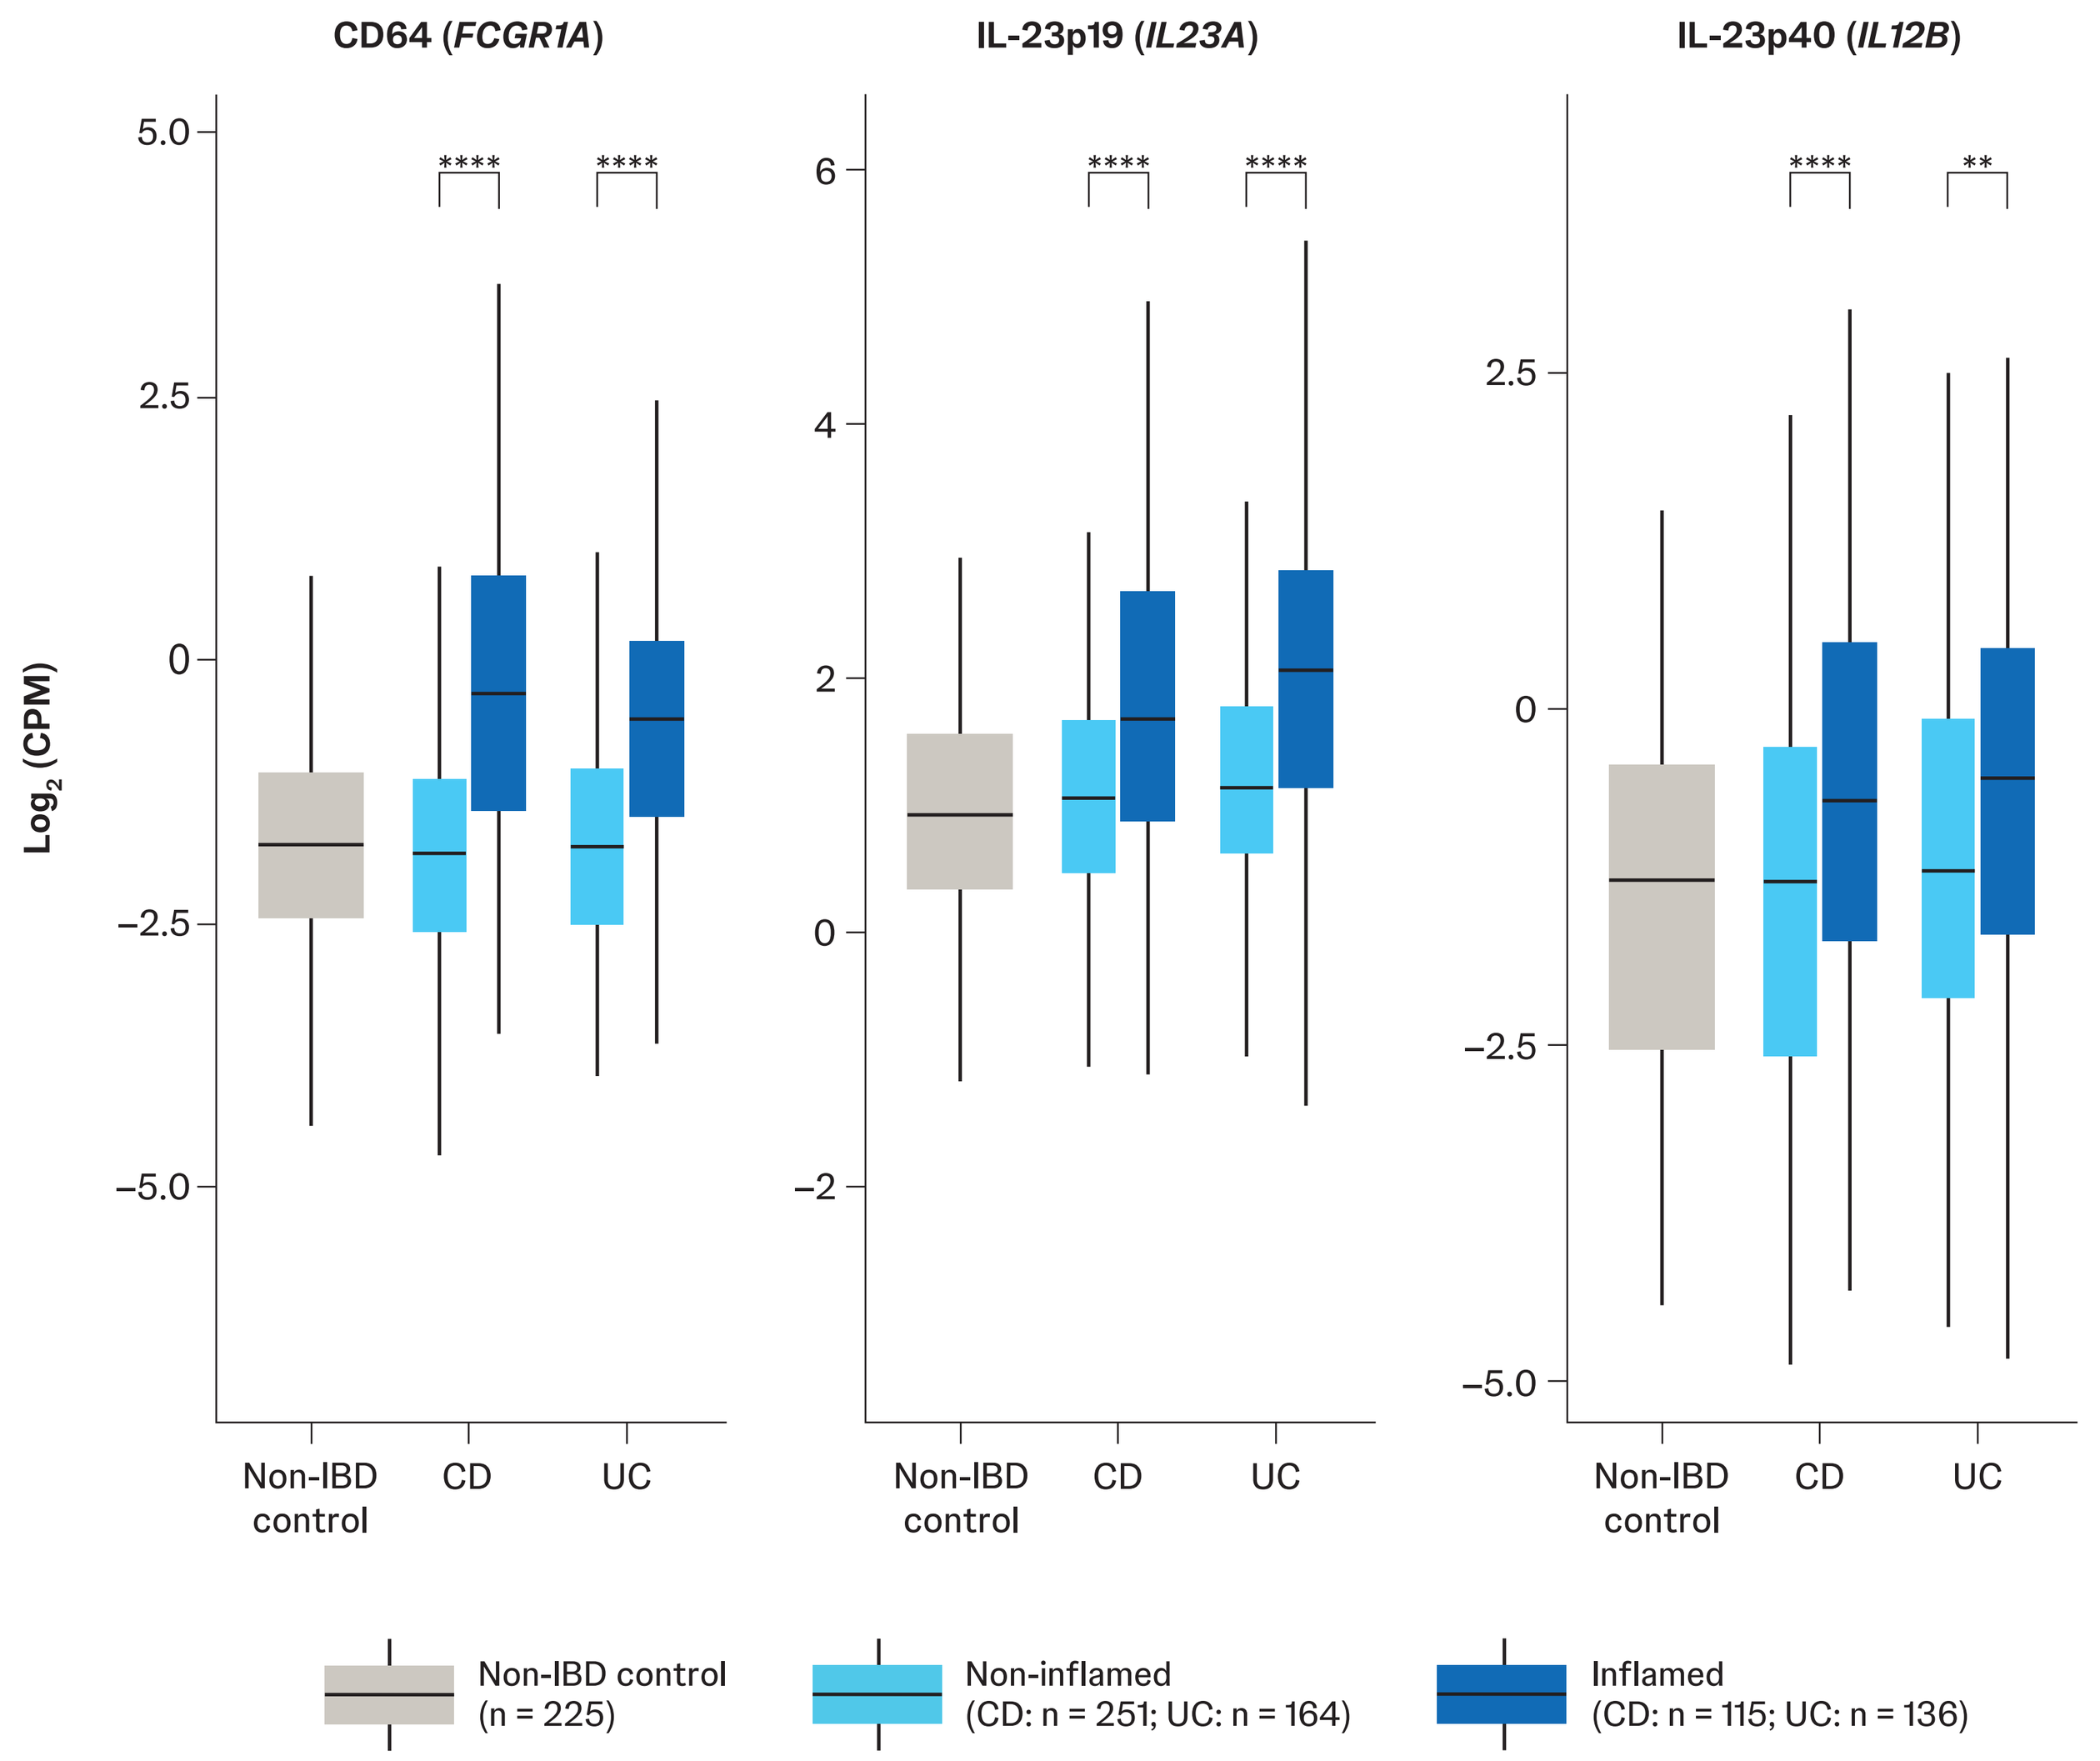
CD64 (FCGR1A)
IL-23p19 (IL23A)
IL-23p40 (IL12B)
Log2 (CPM)
Non-IBD control
CD
UC
Non-IBD control (n = 225)
Non-inflamed (CD: n = 251; UC: n = 164)
Inflamed (CD: n = 115; UC: n = 136)

All comparisons were evaluated with a 2-sample t test (**P <0.01; ****P <0.0001).

Figure 2. *FCGR1A* (CD64) and *IL23A* (IL-23p19) expression from single-cell RNAseq datasets of ileum biopsies from patients with CD (n = 9)

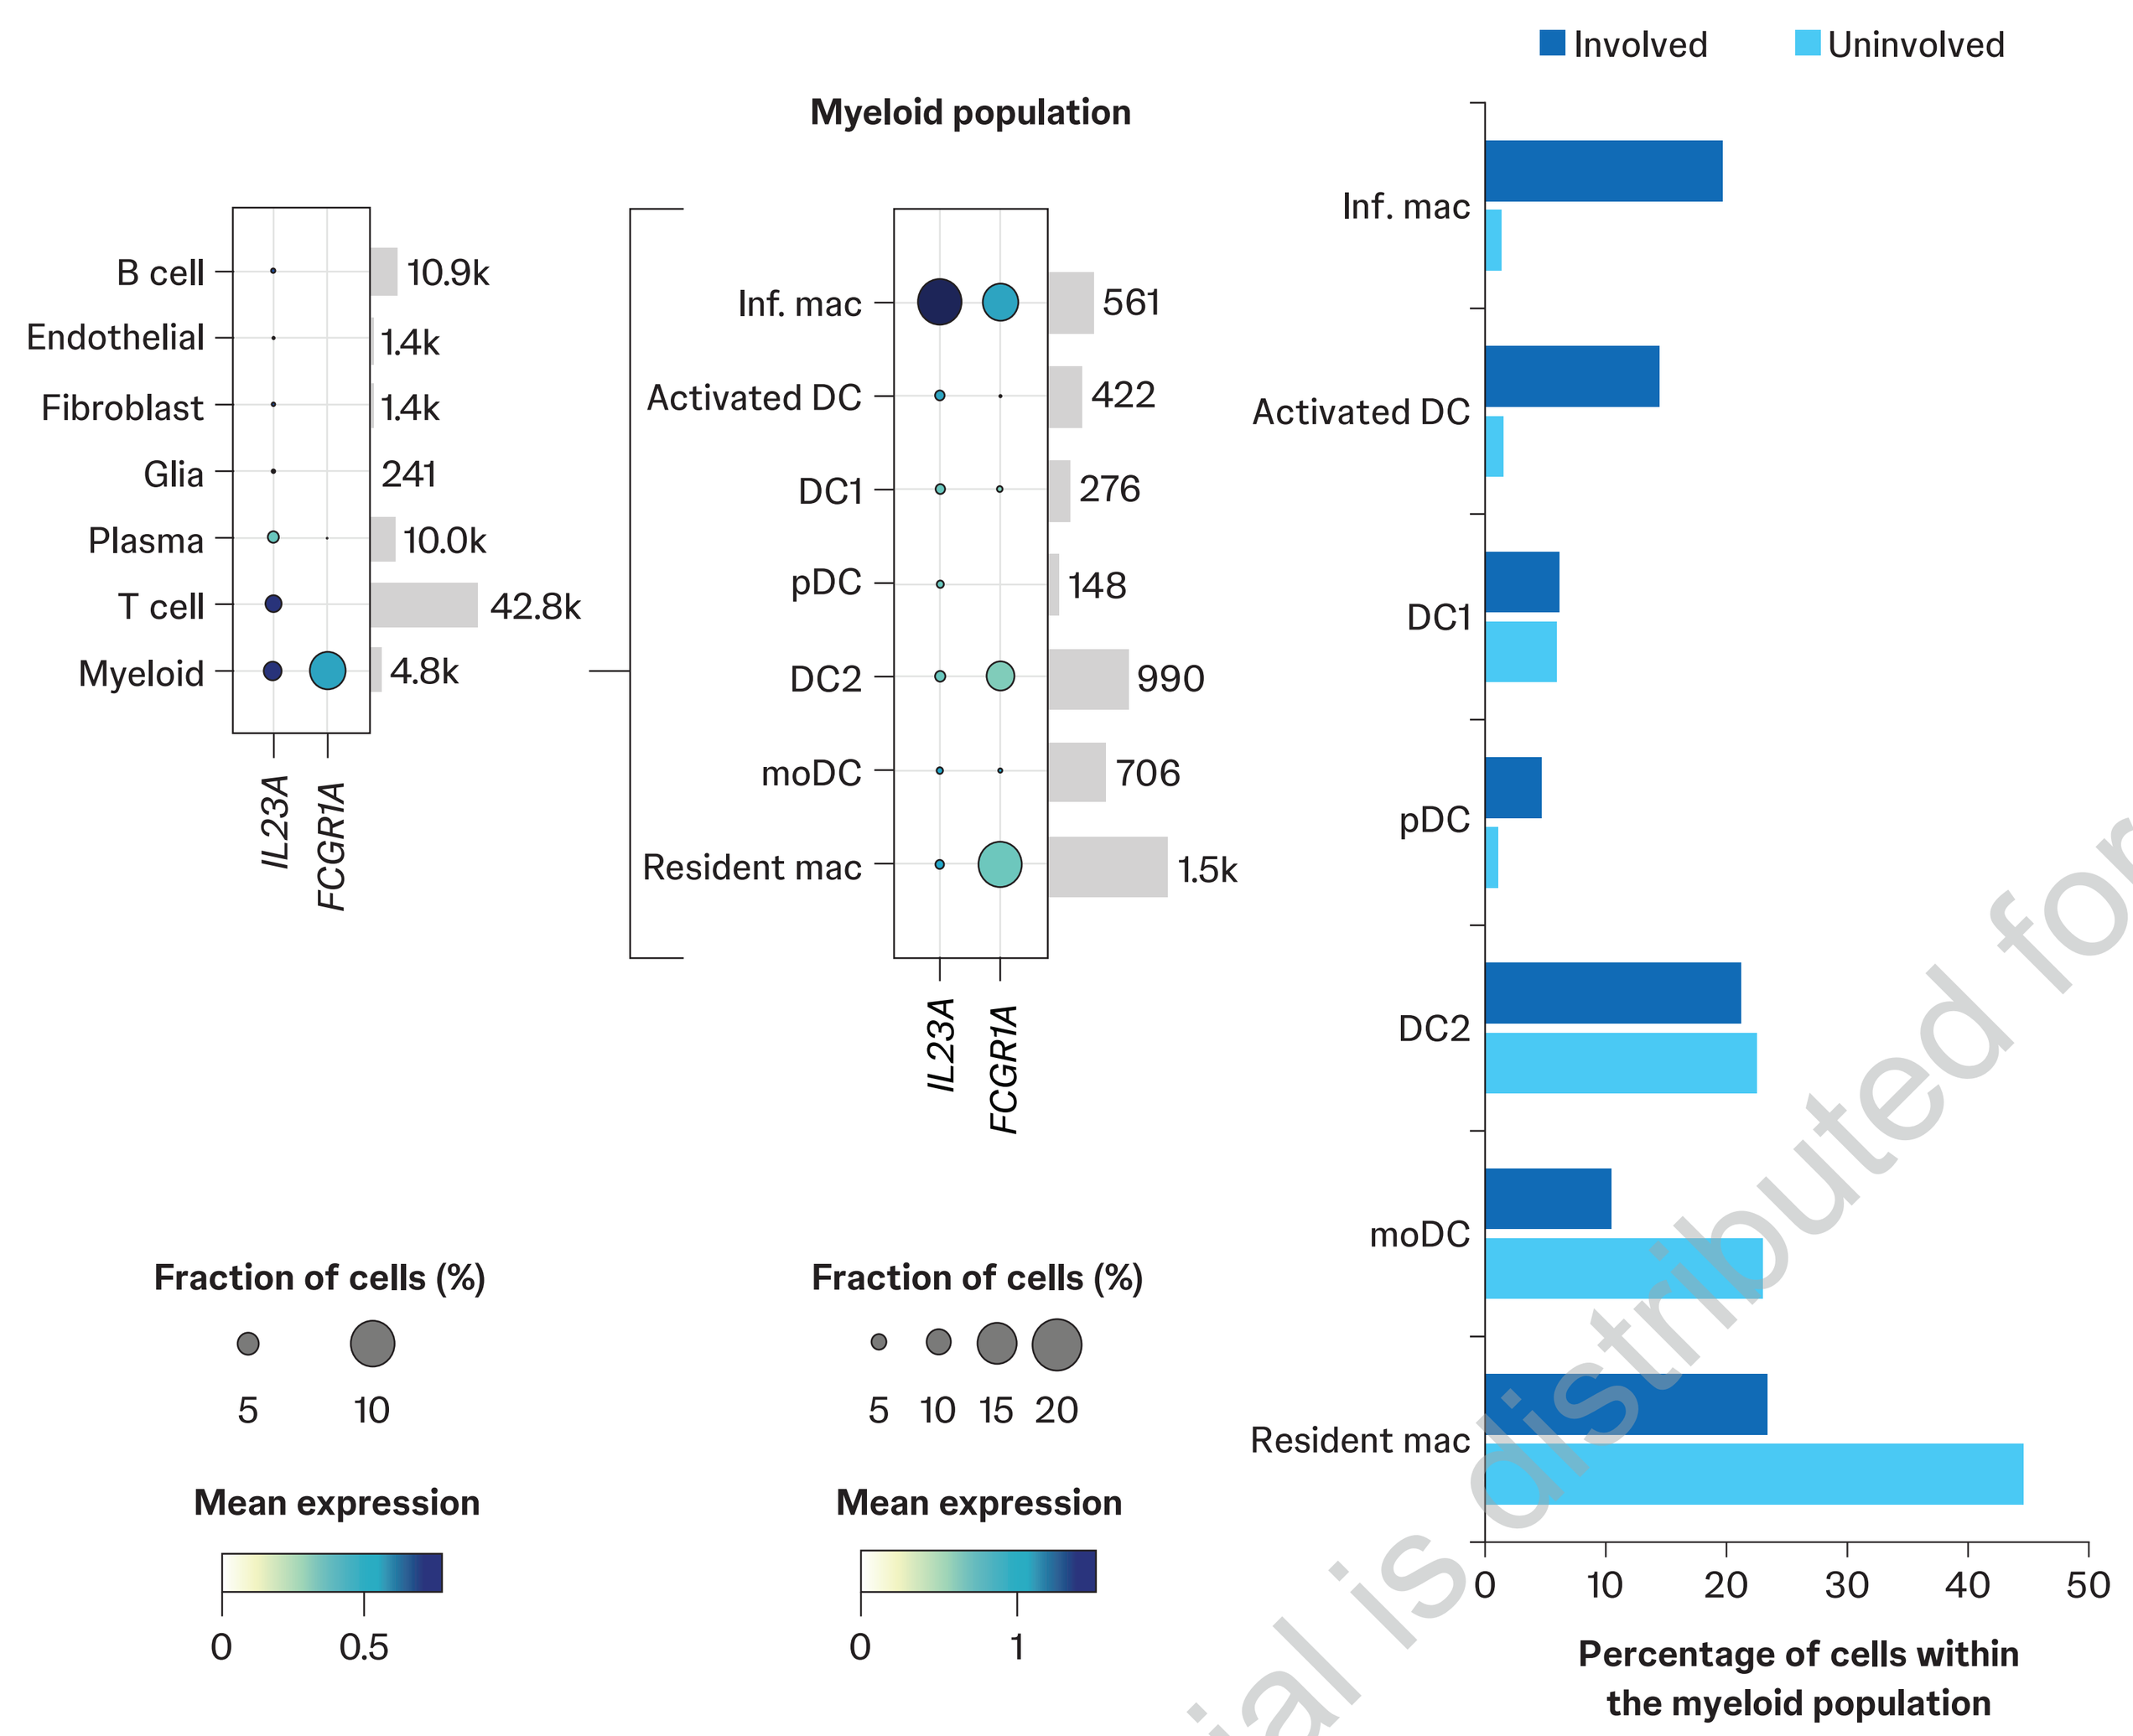
Myeloid population
B cell 10.9k
Endothelial 1.4k
Fibroblast 1.4k
Glia 241
Plasma 10.0k
T cell 42.8k
Myeloid 4.8k
Inf. mac 561
Activated DC 422
DC1 276
pDC 148
DC2 990
moDC 706
Resident mac 1.5k
IL23A
FCGR1A
Fraction of cells (%)
Mean expression
Involved
Uninvolved
Percentage of cells within the myeloid population

Dot plots indicate mean expression level and the fraction of cells expressing IL23A and FCGR1A across various cell types. Cell types are listed to the left of the dot plots; the number of cells of each type in the data used for these analyses is shown on the right. The bar graph shows the percentage of cells in the myeloid population from involved and uninvolved tissues in CD ileum. Inf. mac, inflammatory macrophage; DC, dendritic cell; pDC, plasmacytoid dendritic cell; moDC, monocyte-derived dendritic cell; mac, macrophage.

GUS, RZB, and MIRI displayed similar single-digit picomolar binding affinity for IL-23 (Figure 3)

Figure 3. Binding kinetics and affinity of IL-23p19 subunit mAbs for IL-23 via KinExA

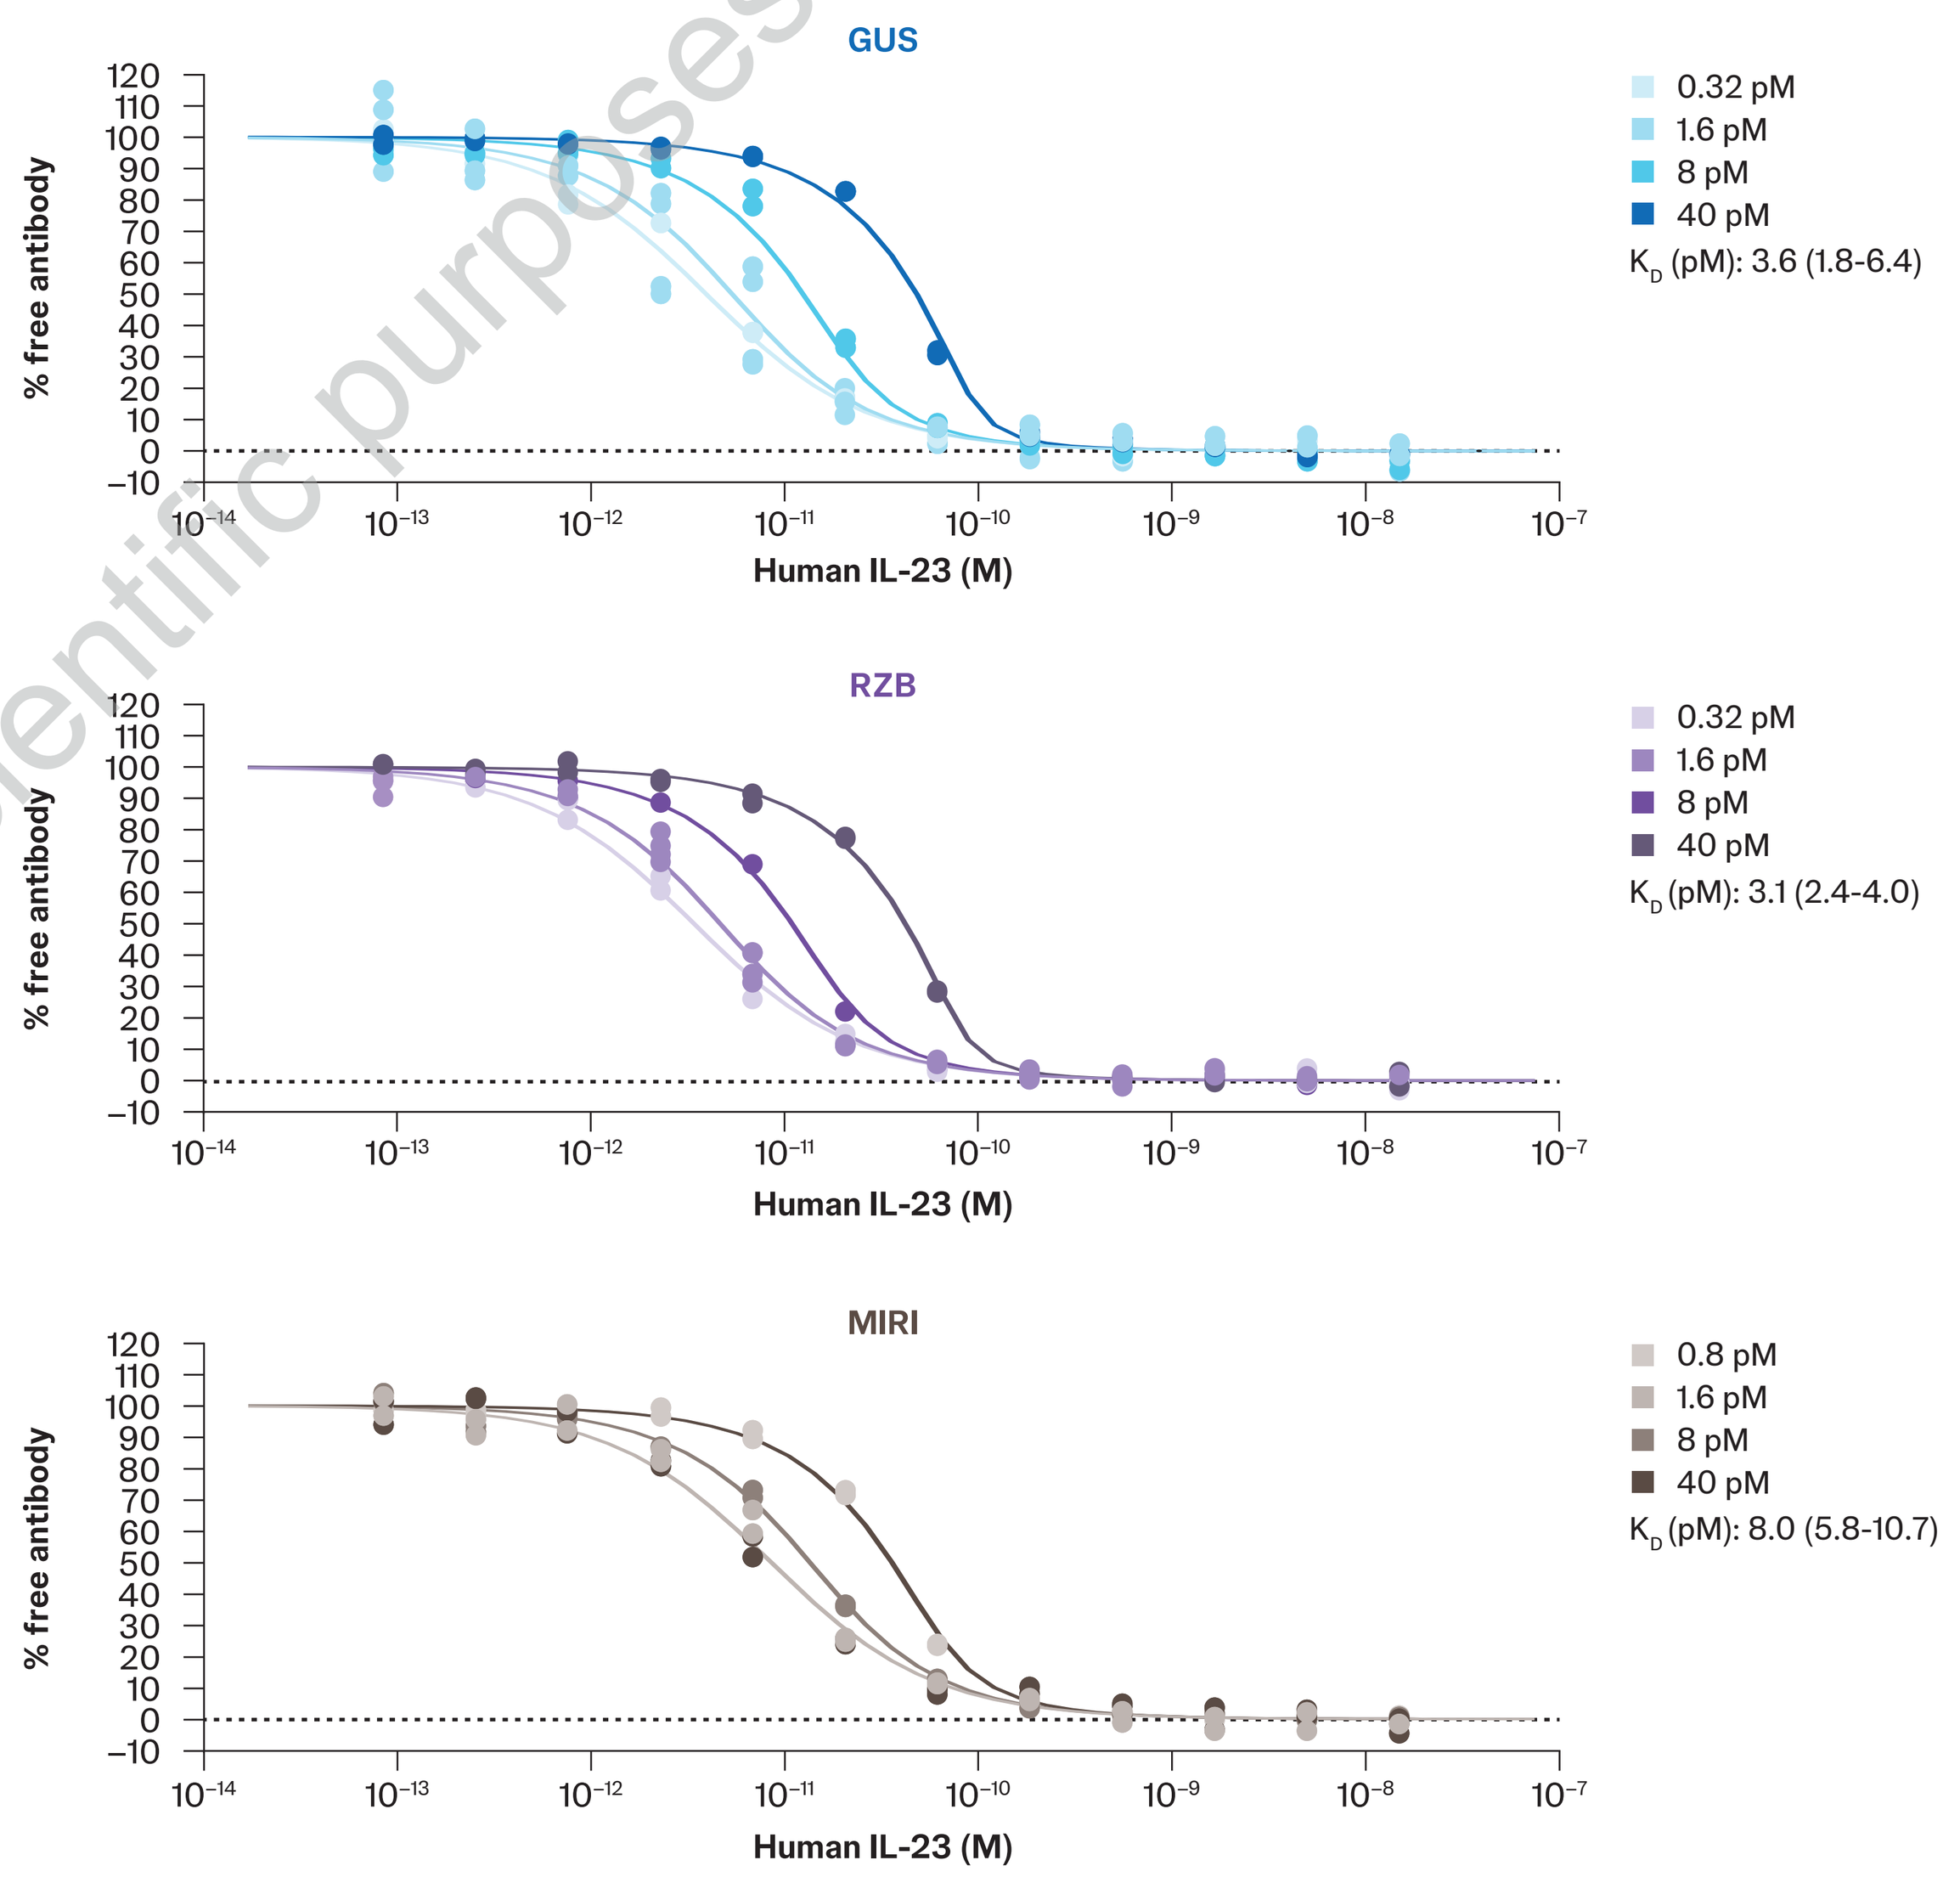
GUS
0.32 pM
1.6 pM
8 pM
40 pM
KD (pM): 3.6 (1.8-6.4)
RZB
0.32 pM
1.6 pM
8 pM
40 pM
KD (pM): 3.1 (2.4-4.0)
MIRI
0.8 pM
1.6 pM
8 pM
40 pM
KD (pM): 8.0 (5.8-10.7)
% free antibody
Human IL-23 (M)

K_D is the result of the global analysis (n-curve analysis) of 4 different antibody concentrations performed to obtain the affinities and the 95% CIs. CI, confidence interval; K_D, equilibrium dissociation constant.

GUS binds to CD64, while no binding was observed for RZB or MIRI (Figure 4)

Figure 4. Evaluation of mAb binding to CD64 by HTRF assay

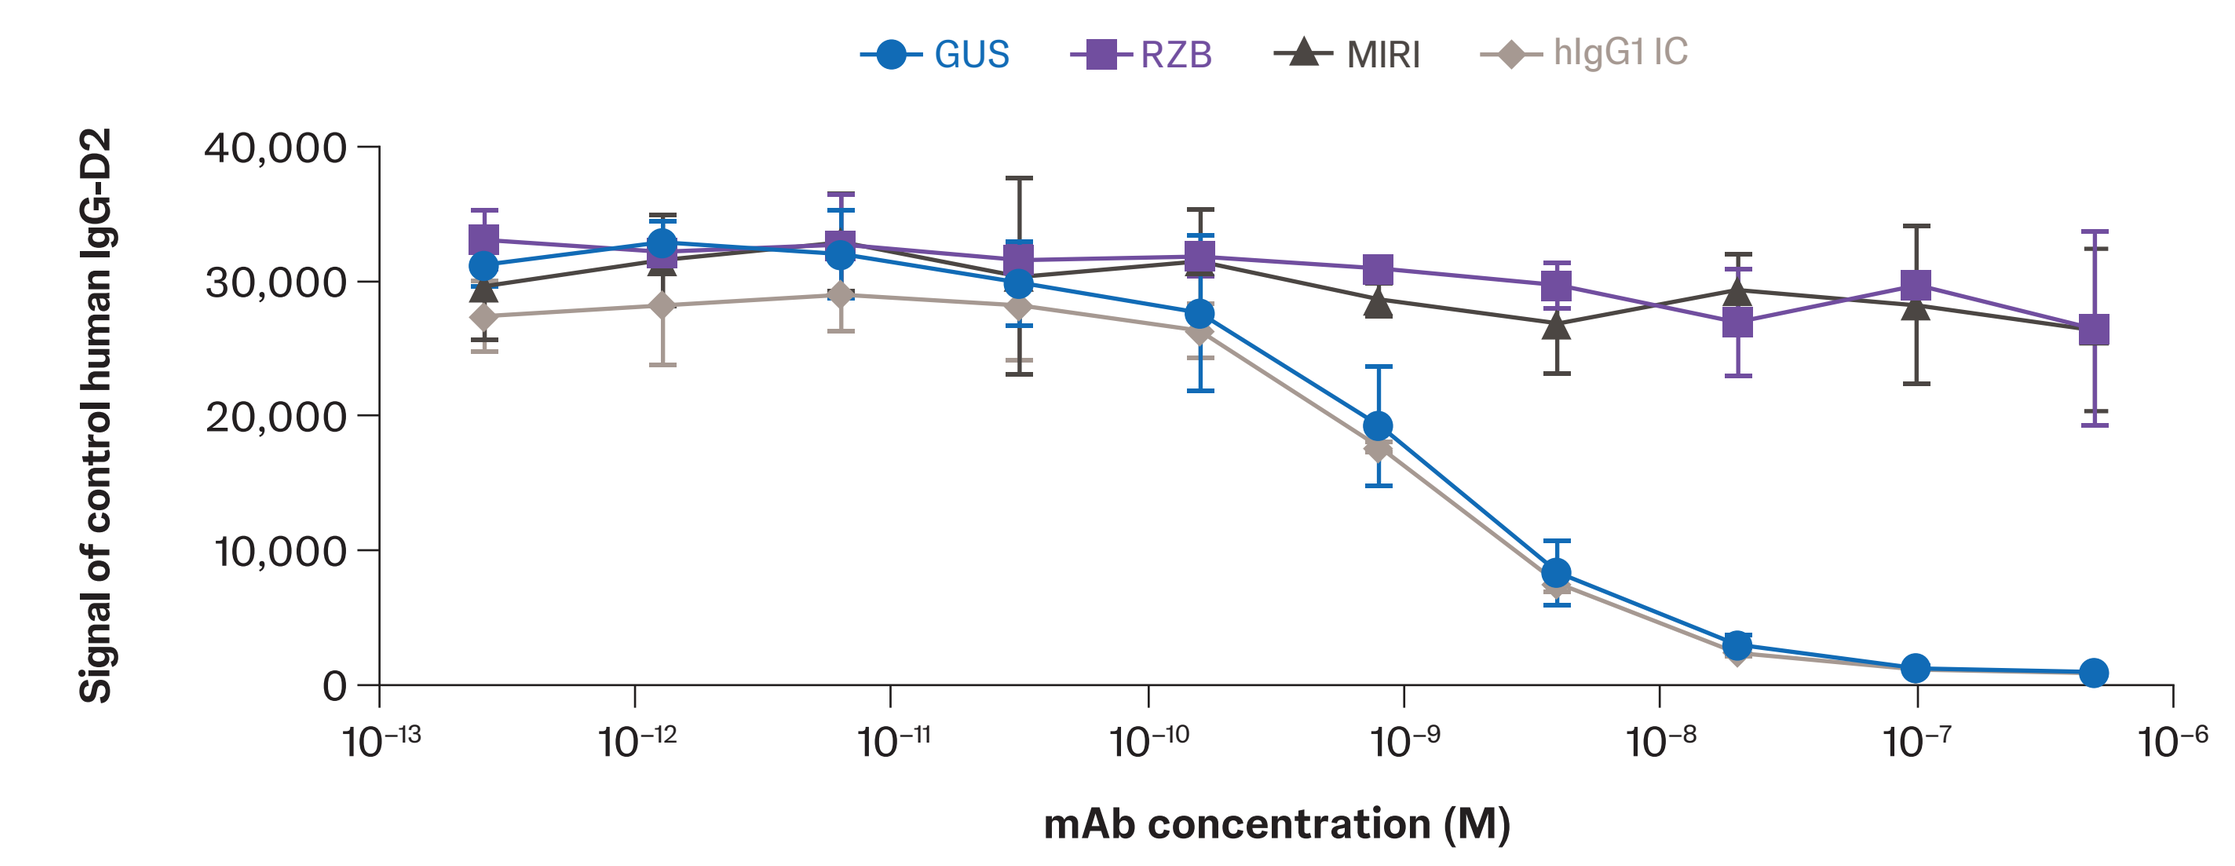
GUS
RZB
MIRI
hIgG1 IC
Signal of control human IgG-D2
mAb concentration (M)

Data are representative of n ≥3 experiments.

GUS, RZB, and MIRI demonstrated comparable potency for inhibition of signaling induced by rhIL-23 in human PBMCs (Figure 5)

Figure 5. Potency for inhibition of rhIL-23 in PBMCs by pSTAT3 assay

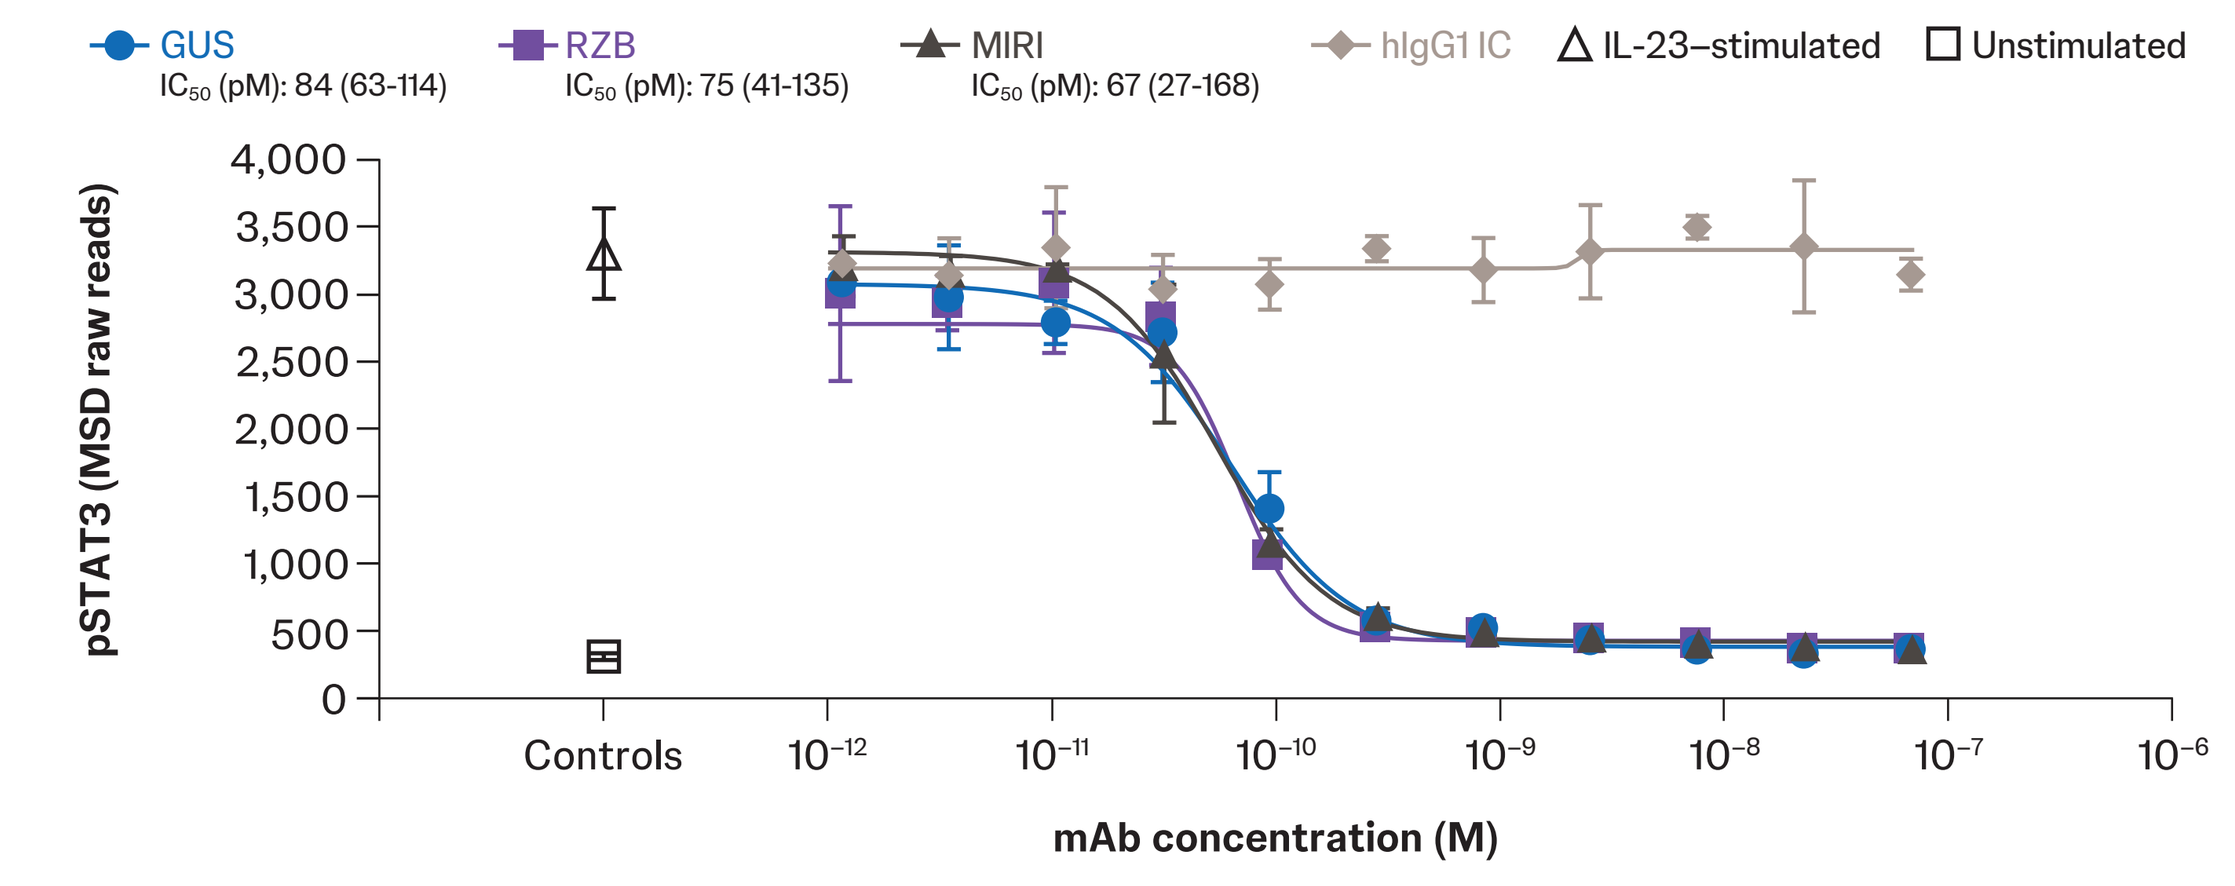
GUS IC50 (pM): 84 (63-114)
RZB IC50 (pM): 75 (41-135)
MIRI IC50 (pM): 67 (27-168)
hIgG1 IC
IL-23–stimulated
Unstimulated
pSTAT3 (MSD raw reads)
Controls
mAb concentration (M)

Plotted data are representative of 3 independent experiments. IC_{50} values (95% CIs) are averaged from 3 independent experiments. IC_{50}, half-maximal inhibitory concentration.

GUS, RZB, and MIRI demonstrated comparable potency for inhibition of signaling with native IL-23 in IL-23 bioassay cells (Figure 6)

Figure 6. Potency for inhibition of exogenous native IL-23 signaling in bioassay cells

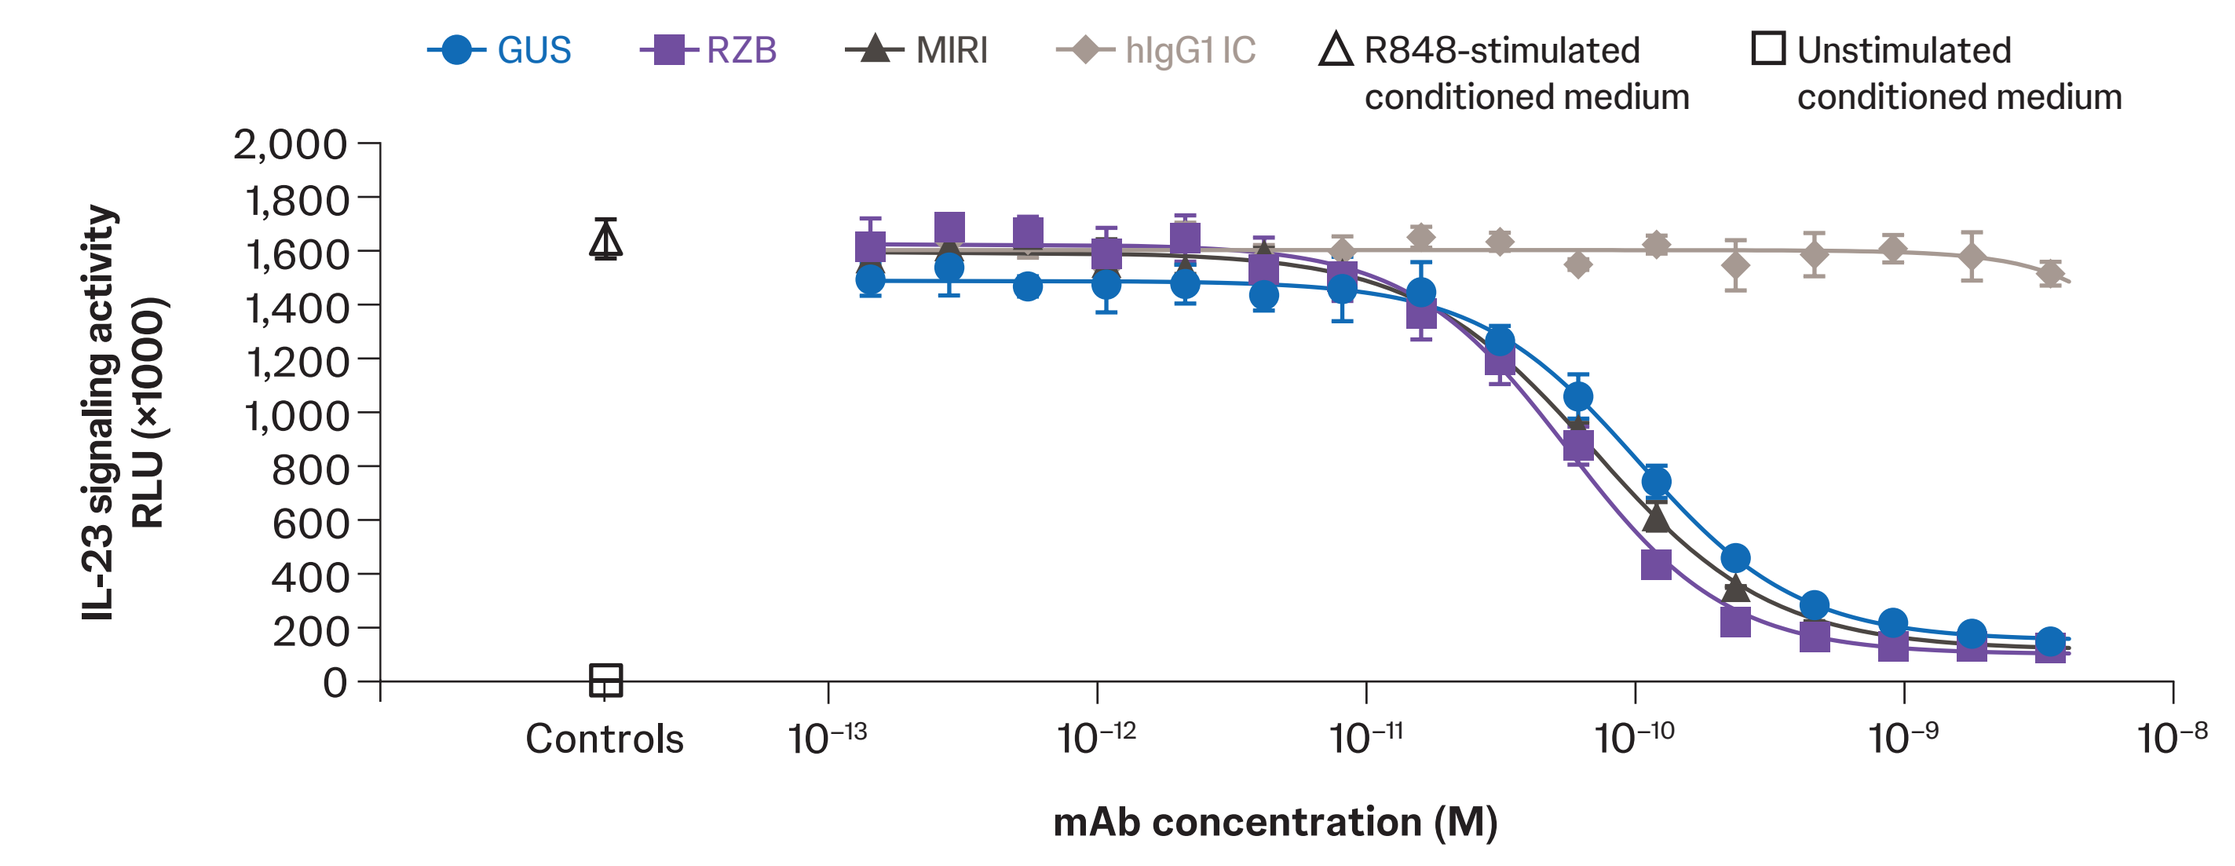
GUS
RZB
MIRI
hIgG1 IC
R848-stimulated conditioned medium
Unstimulated conditioned medium
IL-23 signaling activity RLU (×1000)
Controls
mAb concentration (M)

Plotted data are representative of 3 independent experiments.

GUS displayed enhanced potency compared with RZB and MIRI for neutralization of IL-23 signaling in a co-culture of IL-23–producing CD64⁺ myeloid cells with IL-23–responsive cells (Figure 7)

Figure 7. Potency for inhibition of IL-23 produced locally by CD64⁺ monocytes

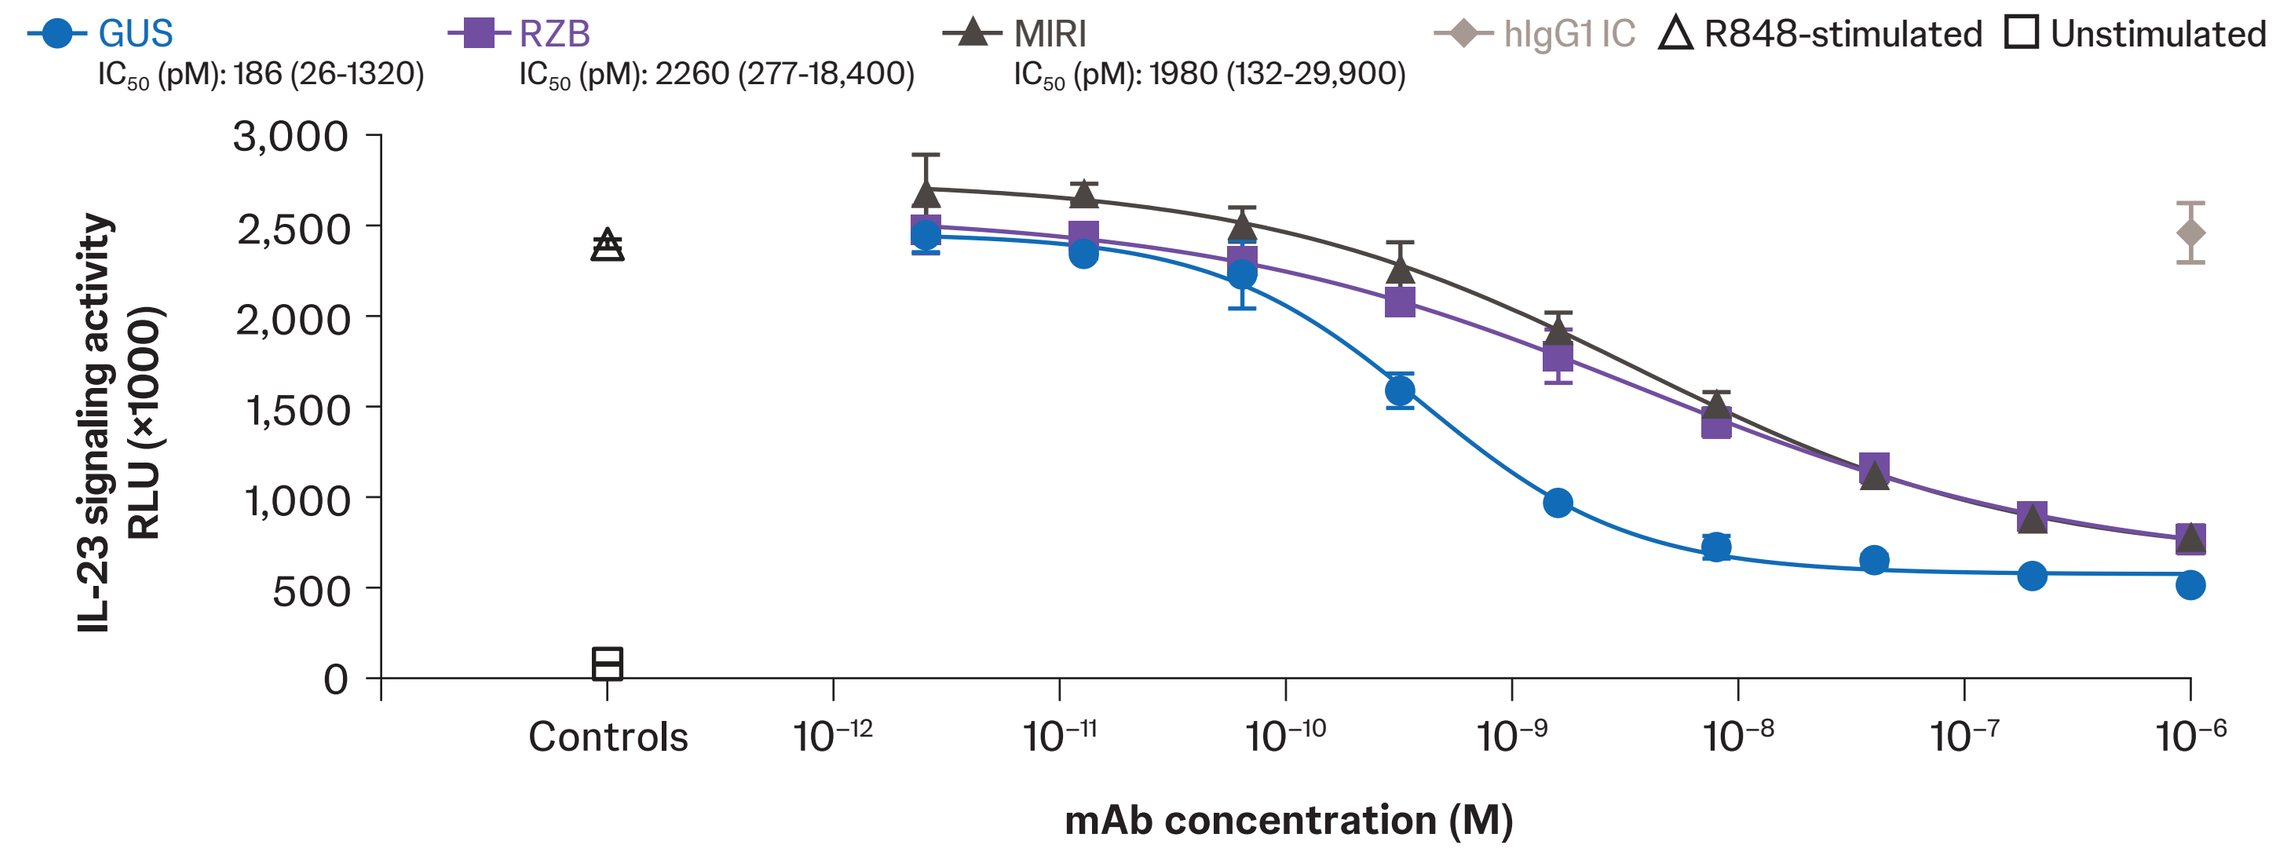
GUS IC50 (pM): 186 (26-1320)
RZB IC50 (pM): 2260 (277-18,400)
MIRI IC50 (pM): 1980 (132-29,900)
hIgG1 IC
R848-stimulated
Unstimulated
IL-23 signaling activity RLU (×1000)
Controls
mAb concentration (M)

Plotted data are representative of 4 independent experiments. Average IC_{50} values (95% CIs) across 4 independent experiments are shown.

PRESENTED AT: American College of Gastroenterology (ACG) Annual Scientific Meeting; Philadelphia, PA, USA; October 25-30, 2024.

REFERENCES: 1. Chapuy L, et al. *J Crohns Colitis.* 2020;14(1):79-95. **2.** Chapuy L, et al. *Mucosal Immunol.* 2019;12(3):703-719. **3.** TREMFYA® (guselkumab) injection, for subcutaneous use [full prescribing information]. Janssen Biotech, Inc.; 2022. **4.** SKYRIZI® (risankizumab-rzaa) injection, for subcutaneous use [full prescribing information]. AbbVie, Inc.; 2022. **5.** Dumet C, et al. *MAbs.* 2019;11(8):1341-1350. **6.** Steere B, et al. *J Pharmacol Exp Ther.* 2023;387(2):180-187. **7.** OMVOH (mirikizumab-mrkz) injection, for intravenous or subcutaneous use [package insert]. Eli Lilly and Company; 2023. **8.** Panaccione R, et al. Presented at: Digestive Disease Week (DDW); Washington, DC, USA; May 19-21, 2024. Abstract #1057b. **9.** Rubin D, et al. Presented at: Digestive Disease Week (DDW); Washington, DC, USA; May 19-21, 2024. Abstract #759. **10.** Argmann C, et al. *Gut.* 2023;72(7):1271-1287. **11.** Robinson MD, et al. *Bioinformatics.* 2010;26(1):139-140. **12.** Martin JC, et al. *Cell.* 2019;178(6):1493-1508.e20. **13.** Wolf FA, et al. *Genome Biol.* 2018;19(1):15.

ACKNOWLEDGMENTS: This study was funded by Janssen Research & Development, LLC. Medical writing support was provided by Panita M. Trenor, PhD, of Lumanity Communications Inc., and was funded by Janssen Global Services, LLC.

DISCLOSURES: JRA: served as a consultant for AbbVie, Adiso, Ferring, Finch, Iterative Scopes, Janssen, Merck, Pfizer, Roivant, and Seres Therapeutics; served as a speaker for AbbVie, Bristol Myers Squibb, and Janssen; and received research support from Janssen, Merck, and Pfizer. **RA:** served as a paid or unpaid consultant for or received honoraria from AbbVie, Amgen, Arena Pharmaceuticals, Biogen, Boehringer Ingelheim, Bristol Myers Squibb, Celgene, Celltrion Healthcare, Dr. Falk Pharma, Ferring, Fresenius Kabi, Galapagos, Gilead, GSK, InDex Pharmaceuticals, Janssen, Kiniksa Pharmaceuticals, Merck Sharp & Dohme, Novartis, Pfizer, Roche, Samsung Bioepis, Stelic, Sterna Biologicals, Takeda, and Tillotts. **MTA:** is a consultant or served on advisory boards for AbbVie, Arena Pharmaceuticals (now Pfizer), Bristol Myers Squibb, Celsius Therapeutics, Gilead, Janssen Global Services, Janssen Pharmaceuticals, Lilly, Pfizer, Prometheus Biosciences, and UCB; and received fees for lecturing from Alimentiv, Janssen Pharmaceuticals, Prime, and WebMD Global LLC. **AH, HL, TCF, EL, MD, PB, TS, JW, IS, KK, AF, and KS:** are current or former employees of Janssen Research & Development, LLC, and may hold stock in Johnson & Johnson.