UNISUS study design: an ongoing, event-driven, Phase 3 superiority study evaluating macitentan 75 mg versus 10 mg in patients with pulmonary arterial hypertension (PAH)

Vallerie V. McLaughlin,[1] Yuichi Tamura,[2] Marc Humbert,[3] Arthur Backer,[4] Neli Boyanova,[4] Hilke Kracker,[4] Cheryl Lassen,[4] Anna Larbalestier,[4] Lilian Sanna,[4] Gurinderpal Doad,[5] Ioana R. Preston,[6] Marius Hoeper[7]

[1]University of Michigan, Ann Arbor, MI, United States; [2]Department of Cardiology, International University of Health and Welfare School of Medicine, Tokyo, Japan; [3]Université Paris-Saclay, Hôpital Bicêtre, Assistance Publique Hôpitaux de Paris, Le Kremlin-Bicêtre, France; [4]Johnson & Johnson, Allschwil, Switzerland; [5]Johnson & Johnson, Titusville, NJ, United States; [6]Lahey Hospital & Medical Center, Burlington, MA, United States; [7]Hannover Medical School, Hannover, Germany

Background

- Pulmonary arterial hypertension (PAH) is a progressive and life-threatening disease of the pulmonary vasculature.[1] Despite improvements in survival with PAH-specific therapies, there remains an unmet need to further improve long-term outcomes for patients with PAH.[2]
- Macitentan is an oral, dual endothelin receptor (ET_A/ET_B) antagonist (ERA) with proven efficacy at a once-daily dose of 10 mg.[1,3] Macitentan is indicated for the treatment of PAH in adults to reduce risk of disease progression and hospitalization for PAH.[4]
- Analyses of clinical and non-clinical data suggest that a dose of 75 mg might provide further benefit to patients with PAH.[4–6]

Objective

- We describe the design and status of the ongoing UNISUS study, which aims to show superior efficacy of macitentan 75 mg over 10 mg in adults with PAH.

Methods

- UNISUS (NCT04273945) is an ongoing, Phase III, prospective, multicenter, double-blind, double-dummy, randomized, active-controlled, parallel-group, group-sequential, adaptive, event-driven study in adults with PAH.
- Following a 4-week run-in period of open-label macitentan 10 mg (if ERA treatment-naïve or on sub-optimal ERA dose), patients were then randomized 1:1 to receive either oral macitentan 75 mg or 10 mg once-daily (**Figure 1**).
- The initial study population was restricted to patients in WHO functional class II/III, aged 18–75 years.
 - Stable (≥3 months) background PAH-therapy could be maintained, except for ERAs.
- The primary endpoint is time to first morbidity/mortality event (defined as PAH-related hospitalization, PAH progression, or death). Details of the primary and secondary endpoints are provided in **Figure 2**.
 - An independent clinical event committee adjudicates all morbidity/mortality events.
- The study has an adaptive design to ensure safety and enroll a broad population. This includes an unblinded independent data monitoring committee (IDMC) conducting two pre-defined interim reviews for futility and sample-size re-estimation, following evaluation of unblinded safety and tolerability data (**Figure 3**).
- UNISUS has several sub-studies, including a cardiac remodeling sub-study, hemodynamic sub-study, and pharmacokinetic–pharmacodynamic sub-study (**Figure 4**).

Figure 1. UNISUS is an ongoing, multicenter, prospective, Phase III, superiority study of macitentan 75 mg versus 10 mg in adults with PAH

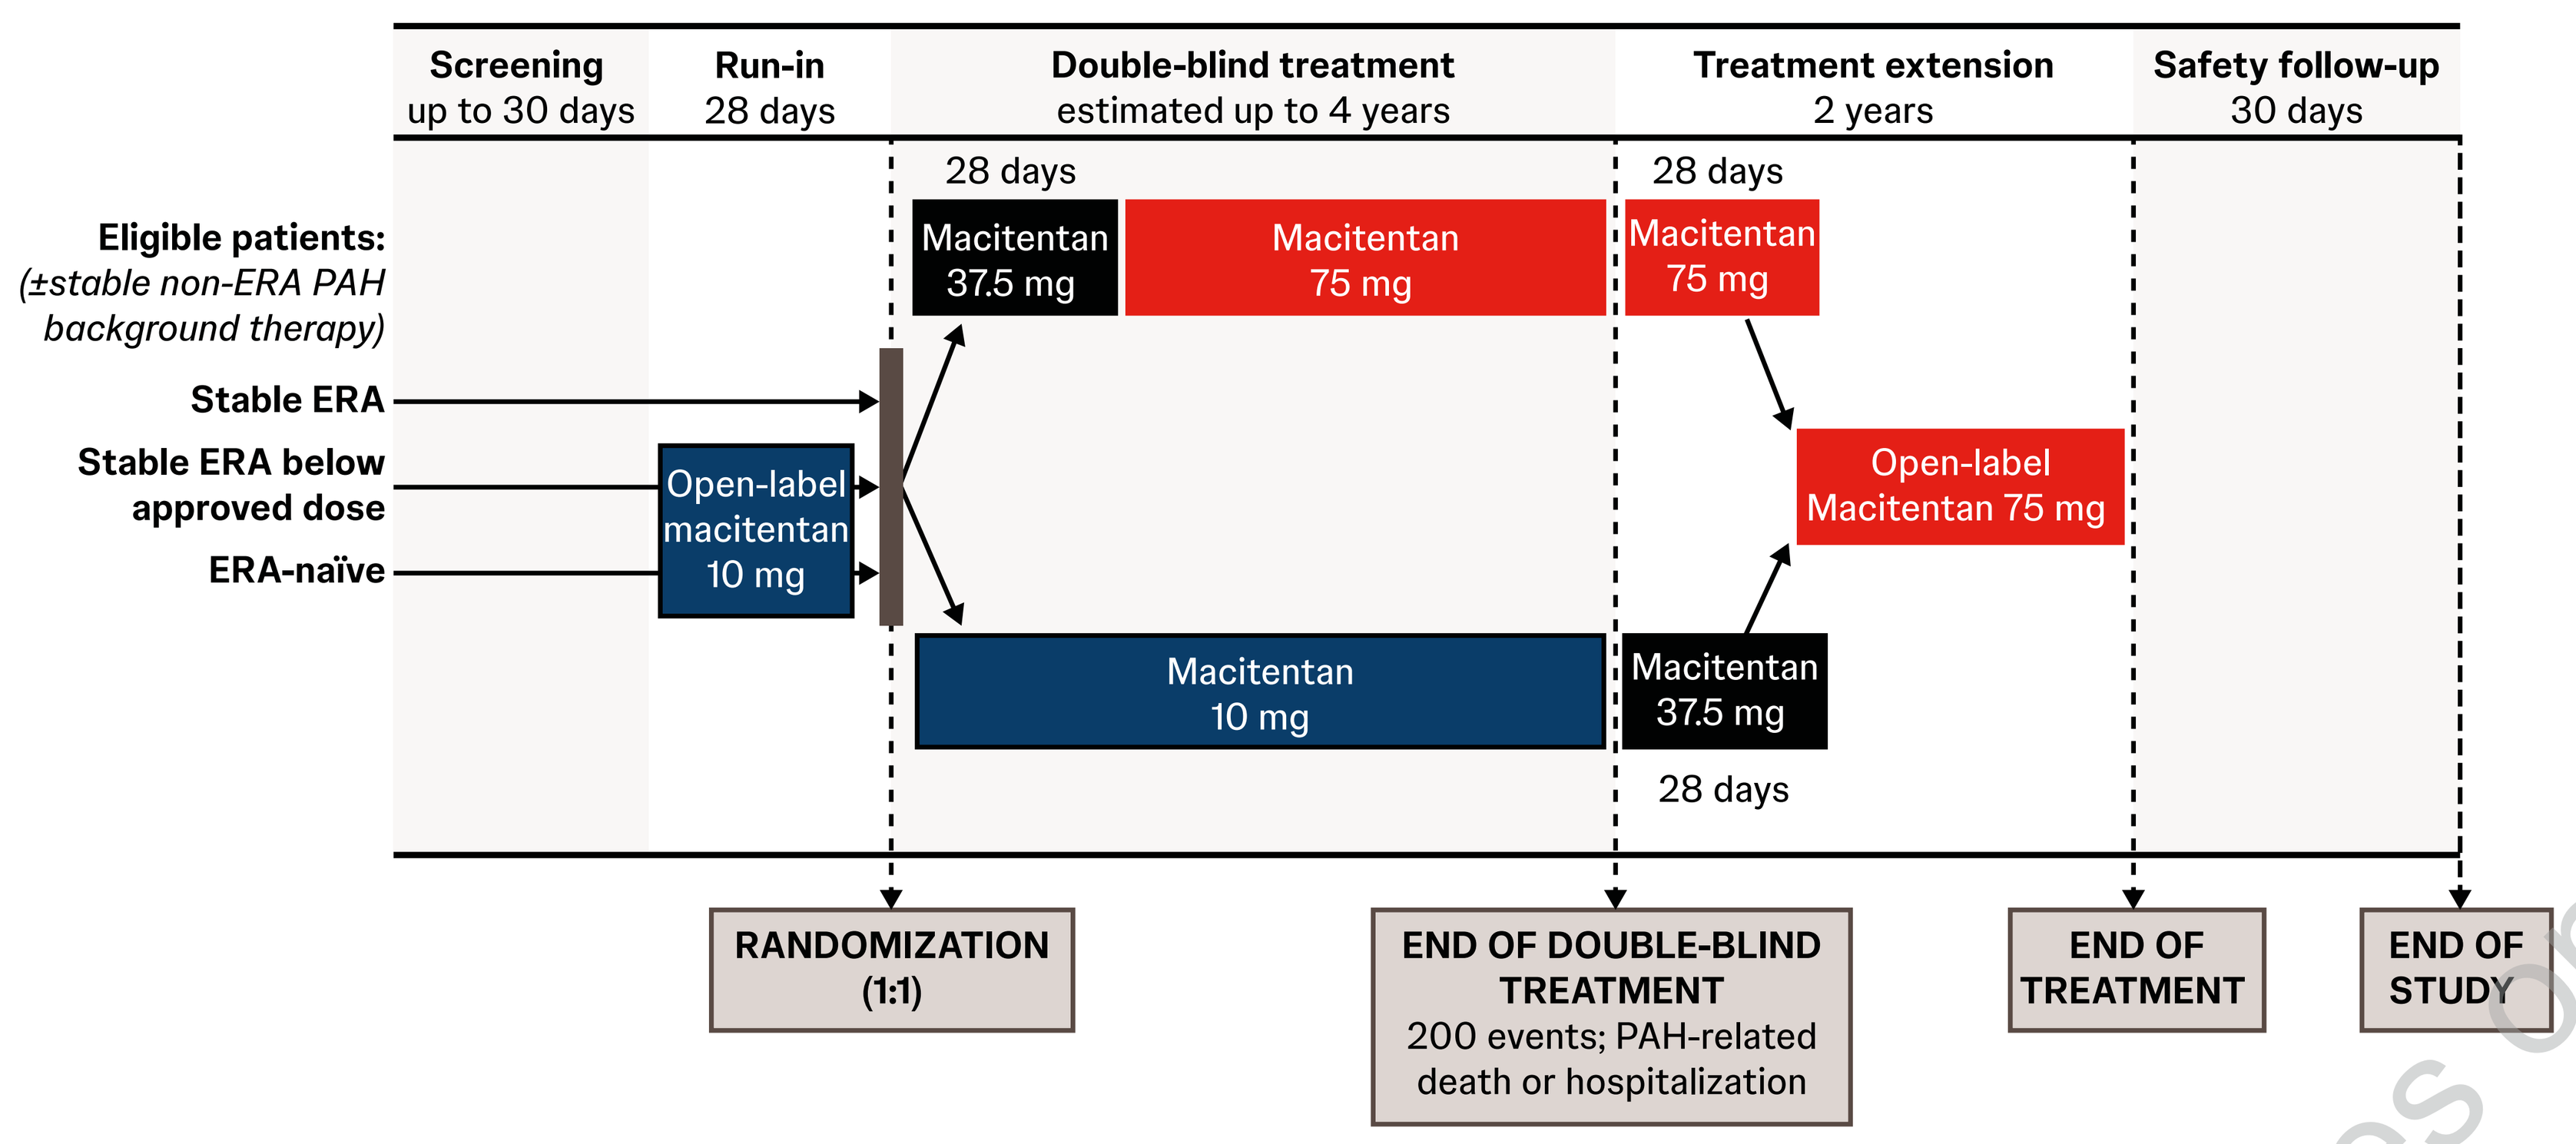
Screening
up to 30 days
Run-in
28 days
Double-blind treatment
estimated up to 4 years
Treatment extension
2 years
Safety follow-up
30 days
Eligible patients:
(±stable non-ERA PAH background therapy)
Stable ERA
Stable ERA below approved dose
ERA-naïve
Open-label macitentan 10 mg
28 days
Macitentan 37.5 mg
Macitentan 75 mg
Macitentan 10 mg
Macitentan 75 mg
Open-label Macitentan 75 mg
Macitentan 37.5 mg
28 days
RANDOMIZATION (1:1)
END OF DOUBLE-BLIND TREATMENT
200 events; PAH-related death or hospitalization
END OF TREATMENT
END OF STUDY

ERA, endothelin receptor agonist; PAH, pulmonary arterial hypertension.
Figure first presented at the ESC congress 2022, Barcelona, Spain.

Figure 2. Primary and secondary endpoints of the UNISUS study

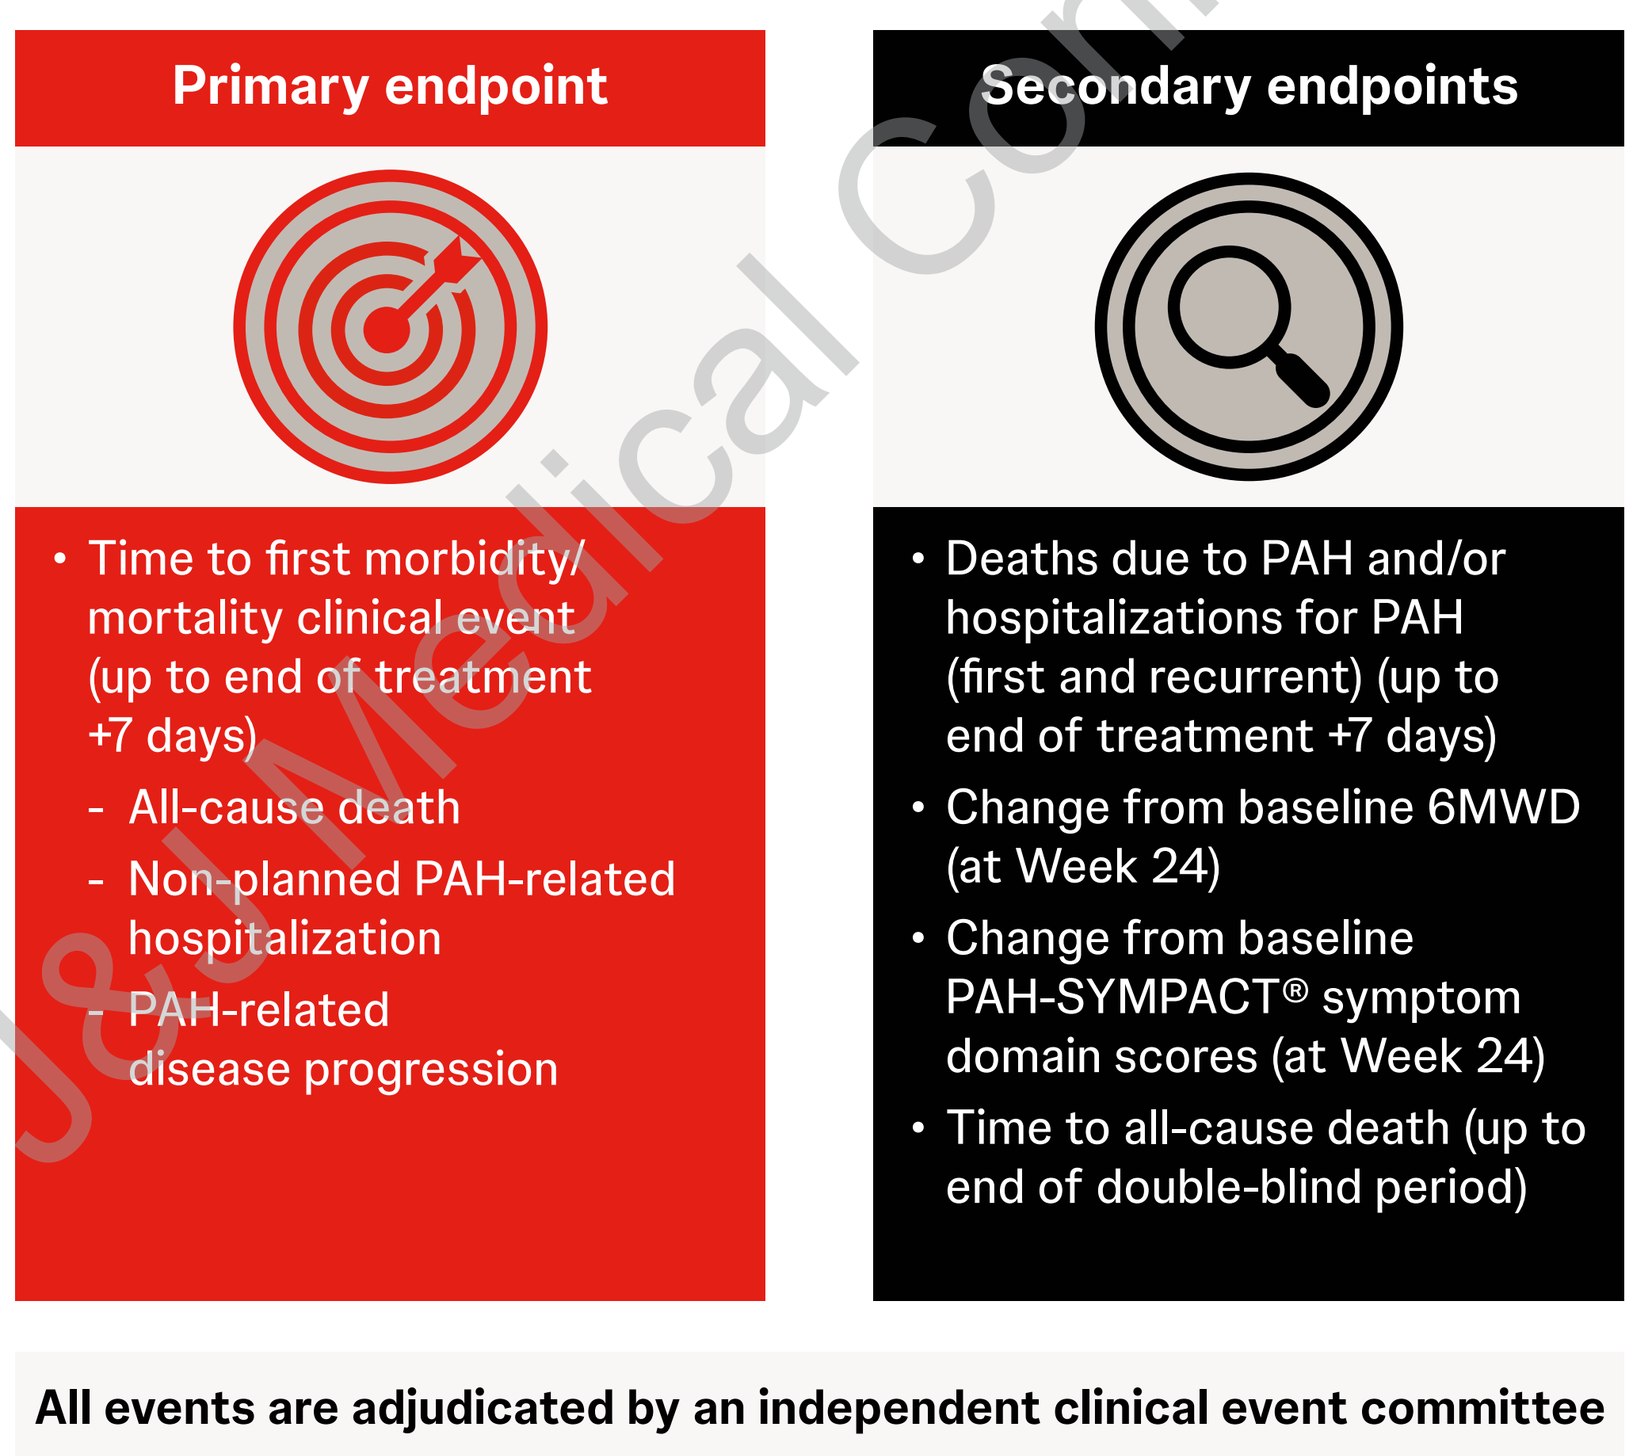
Primary endpoint
• Time to first morbidity/ mortality clinical event (up to end of treatment +7 days)
- All-cause death
- Non-planned PAH-related hospitalization
- PAH-related disease progression
Secondary endpoints
• Deaths due to PAH and/or hospitalizations for PAH (first and recurrent) (up to end of treatment +7 days)
• Change from baseline 6MWD (at Week 24)
• Change from baseline PAH-SYMPACT® symptom domain scores (at Week 24)
• Time to all-cause death (up to end of double-blind period)
All events are adjudicated by an independent clinical event committee

6MWD, 6-minute walk distance; PAH, pulmonary arterial hypertension; PAH-SYMPACT, pulmonary arterial hypertension symptoms and impact.

Figure 3. IDMC-recommended changes following unblinded review of safety/tolerability data and interim futility analysis

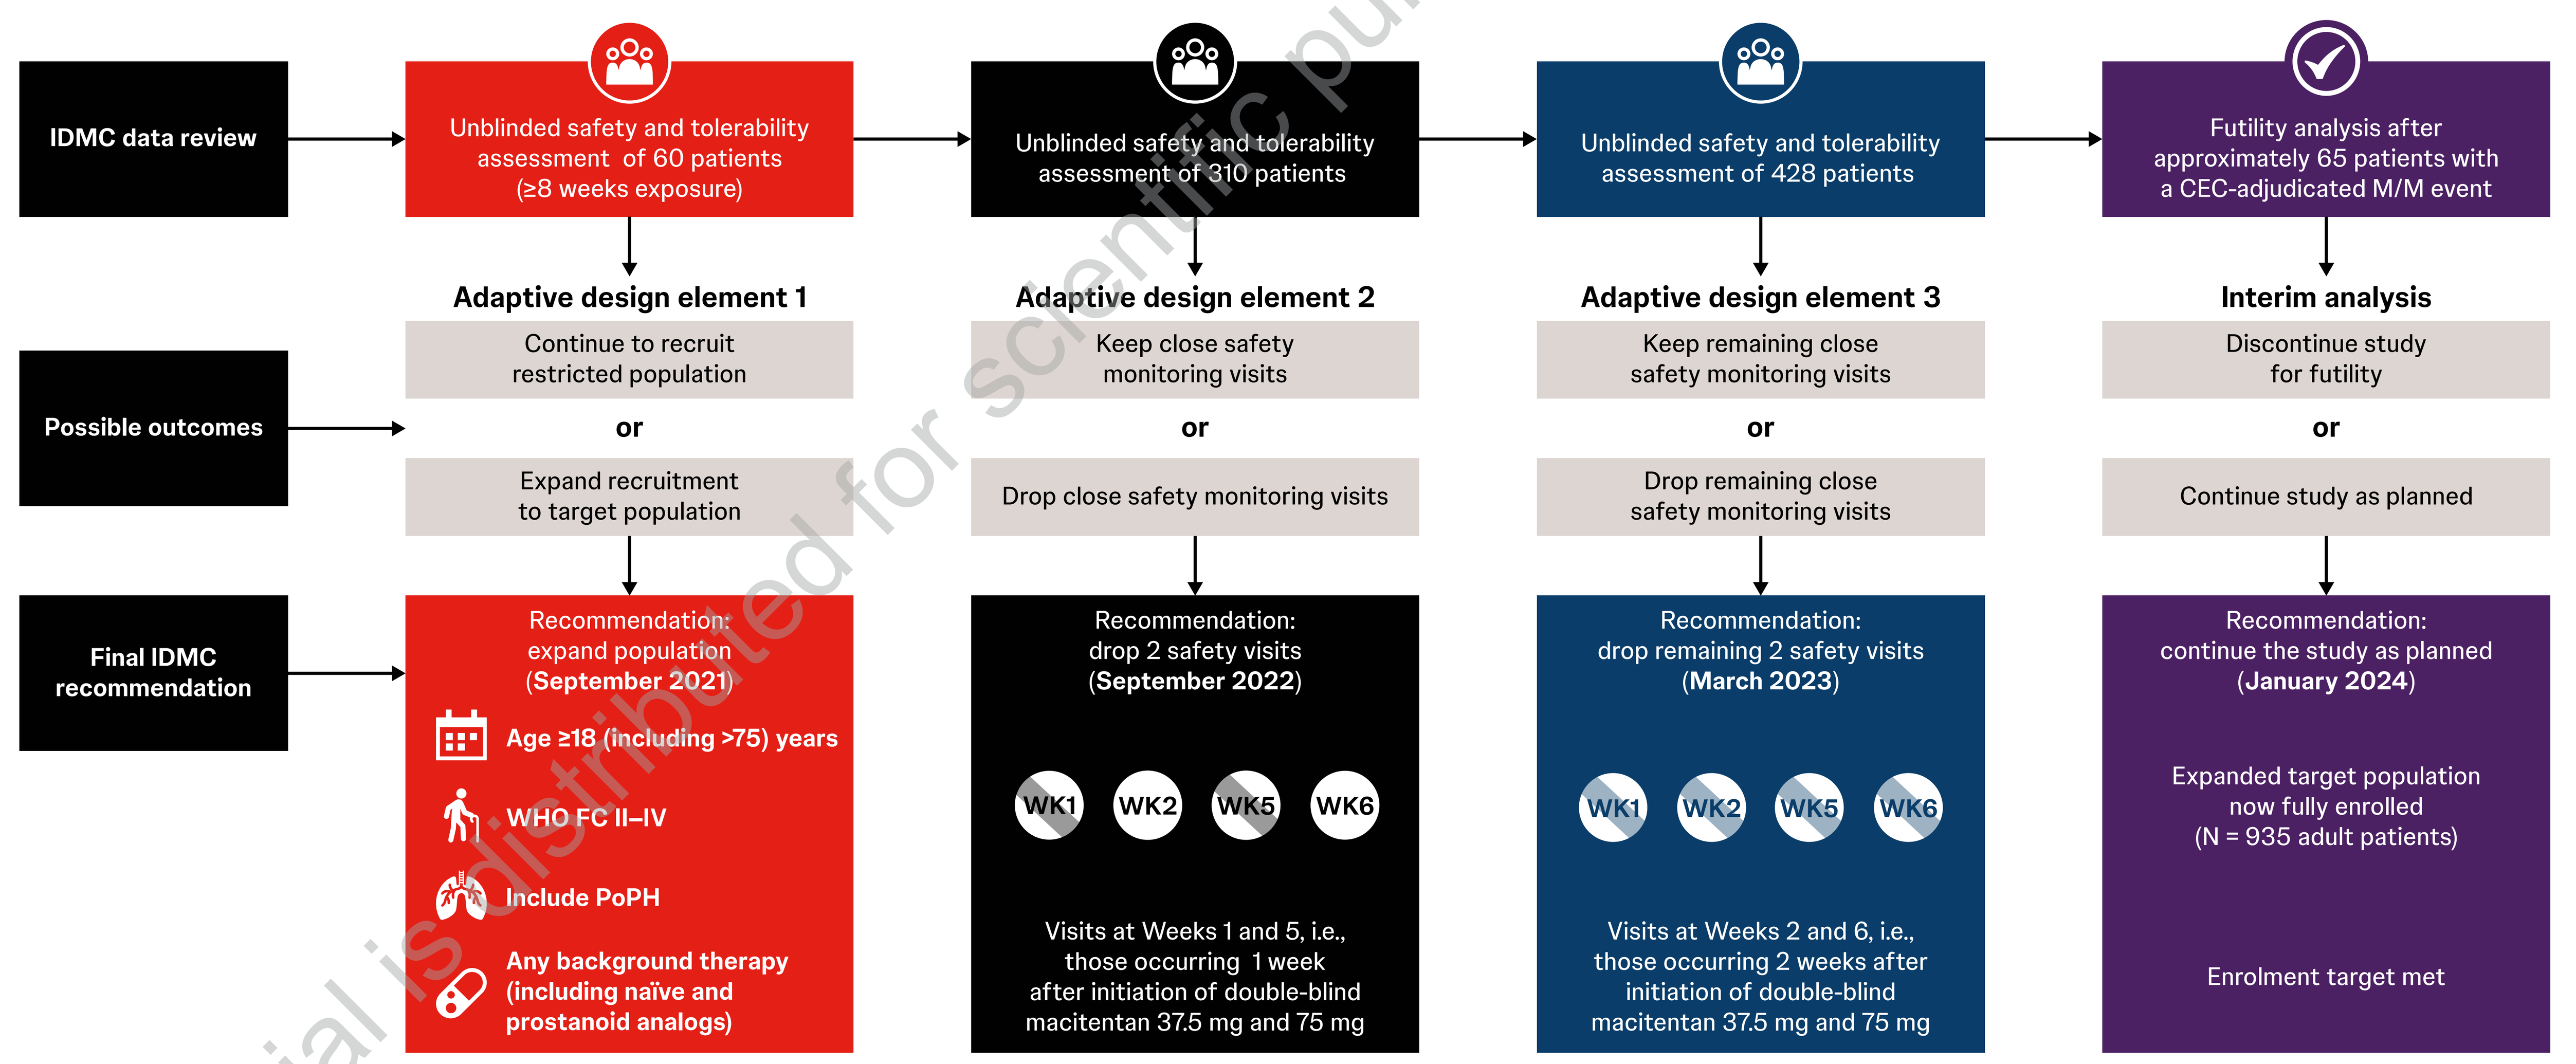
IDMC data review
Possible outcomes
Final IDMC recommendation
Unblinded safety and tolerability assessment of 60 patients (≥8 weeks exposure)
Adaptive design element 1
Continue to recruit restricted population
or
Expand recruitment to target population
Recommendation: expand population (September 2021)
Age ≥18 (including >75) years
WHO FC II–IV
Include PoPH
Any background therapy (including naïve and prostanoid analogs)
Unblinded safety and tolerability assessment of 310 patients
Adaptive design element 2
Keep close safety monitoring visits
or
Drop close safety monitoring visits
Recommendation: drop 2 safety visits (September 2022)
WK1 WK2 WK5 WK6
Visits at Weeks 1 and 5, i.e., those occurring 1 week after initiation of double-blind macitentan 37.5 mg and 75 mg
Unblinded safety and tolerability assessment of 428 patients
Adaptive design element 3
Keep remaining close safety monitoring visits
or
Drop remaining close safety monitoring visits
Recommendation: drop remaining 2 safety visits (March 2023)
WK1 WK2 WK5 WK6
Visits at Weeks 2 and 6, i.e., those occurring 2 weeks after initiation of double-blind macitentan 37.5 mg and 75 mg
Futility analysis after approximately 65 patients with a CEC-adjudicated M/M event
Interim analysis
Discontinue study for futility
or
Continue study as planned
Recommendation: continue the study as planned (January 2024)
Expanded target population now fully enrolled (N = 935 adult patients)
Enrolment target met

CEC, clinical event committee; IDMC, independent data monitoring committee; M/M, morbidity/mortality; PoPH, portopulmonary hypertension; WHO FC, World Health Organization functional class; WK, week.

Figure 4. UNISUS sub-studies

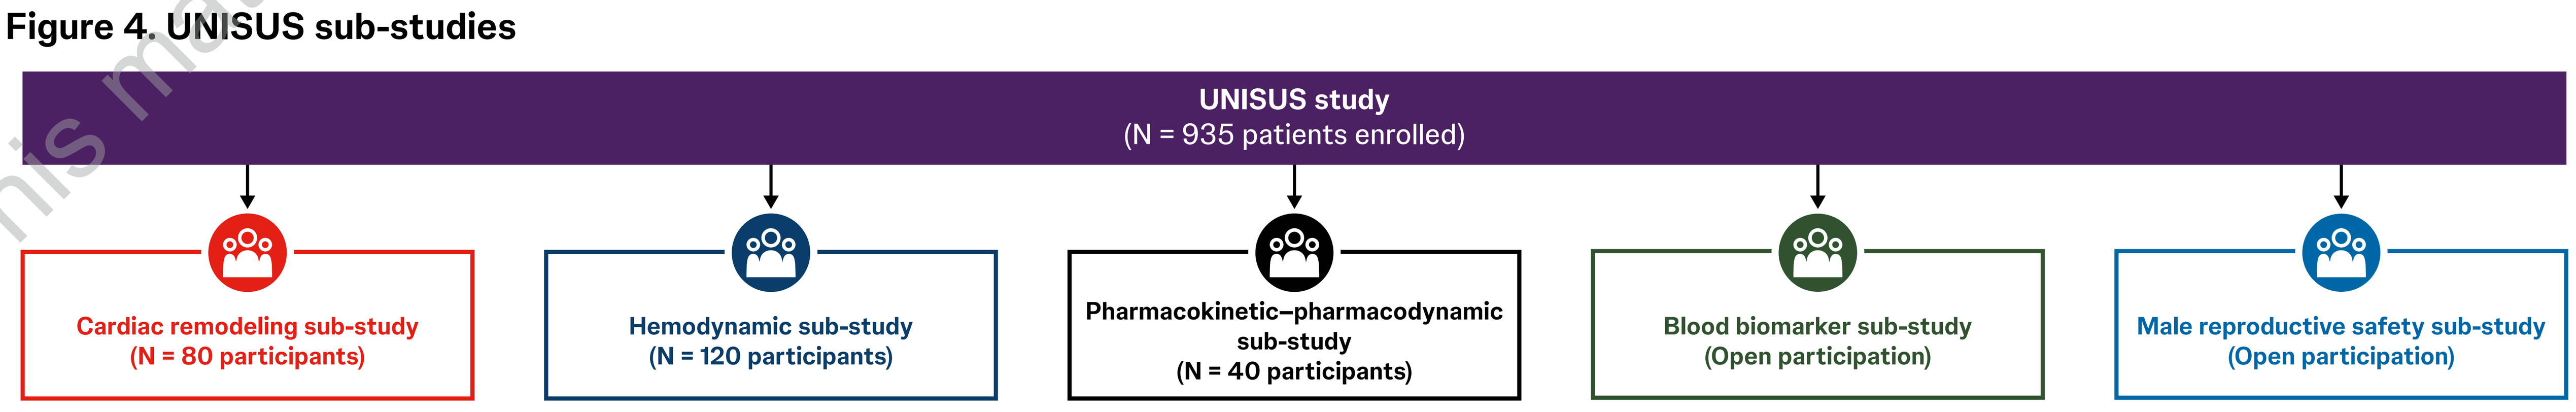
UNISUS study
(N = 935 patients enrolled)
Cardiac remodeling sub-study
(N = 80 participants)
Hemodynamic sub-study
(N = 120 participants)
Pharmacokinetic–pharmacodynamic sub-study
(N = 40 participants)
Blood biomarker sub-study
(Open participation)
Male reproductive safety sub-study
(Open participation)

Status

- Based on unblinded safety and tolerability data from the initial 60 recruited patients with ≥8 weeks of treatment exposure, the IDMC recommended to expand the study population (**Figure 3**) to also include:
 - Patients aged >75 years
 - Patients in WHO functional class IV
 - Patients with portopulmonary hypertension
 - Patients who are treatment-naïve or receiving prostanoid analogs.
- Recruitment to UNISUS is now complete, with 935 adult patients enrolled in the study.

Key takeaway

The ongoing head-to-head UNISUS study, examining if macitentan 75 mg provides superior clinical benefit over the established 10 mg dose, has fully enrolled with 935 adult patients with PAH.

Conclusions

As the first head-to-head superiority study in PAH, UNISUS uses a robust, adaptive, event-driven design to determine if macitentan 75 mg provides superior clinical benefit to patients with PAH over the established 10 mg dose.

Regular unblinded safety reviews of the UNISUS data by the IDMC led to a recommendation of pre-planned study design changes, including:

- An expanded patient population
- Less frequent safety monitoring visits during the up-titration phase.

UNISUS is currently ongoing and has met its enrolment target, with 935 adult patients fully enrolled.

Acknowledgments

Medical writing support was provided by Patrick Hoggard, of Ashfield MedComms, an Inizio company, and funded by Johnson & Johnson.

Disclosures

This study was funded by Johnson & Johnson. Vallerie V. McLaughlin served as a Scientific Committee member for Janssen Pharmaceutical Companies of Johnson & Johnson; received research grants from Aerovate, Altavant, Gossamer Bio, Janssen Pharmaceutical Companies of Johnson & Johnson, Merck and SoniVie; and received consultant fees from Aerami, Aerovate, Altavant, Bayer, Caremark, Corvista, Gossamer Bio, Janssen Pharmaceutical Companies of Johnson & Johnson, L.L.C, Merck, and United Therapeutics. Marius Hoeper has received fees for consultations or lectures from Acceleron, Actelion, Aerovate, AOP Health, Bayer, Ferrer, Gossamer, Janssen, MSD, and Novartis. Arthur Backer, Neli Boyanova, Hilke Kracker, Cheryl Lassen, Anna Larbalestier, Lilian Sanna, and Gurinderpal Doad are employees of Johnson & Johnson.

Posters on UNISUS have been presented at European Cardiology Society (ECS) Congress 2022, American Thoracic Society (ATS) 2024 Congress, and Global Cardiovascular Clinical Trialists forum (CVCT) 2024.

Pulmonary Hypertension

Please scan QR code

Poster

The QR code is intended to provide scientific information for individual reference, and the information should not be altered or reproduced in any way.

Presented by V McLaughlin at Global Cardiovascular Clinical Trialists forum (CVCT); December 8–10, 2025, The Mayflower Hotel, Washington DC, USA.

References

1. Humbert M, et al. *Eur Heart J.* 2022;43:3618–713. 2. Reinders S, et al. *Respir Res.* 2024;25:373. 3. Pulido T, et al. *N Engl J Med.* 2013;369:809–18. 4. Johnson & Johnson. Opsumit®. Highlights of prescribing information. https://www.opsumit.com/opsumit-prescribing-information.pdf (accessed Oct 2025). 5. Csonka D, et al. Poster presented at ERS 2025, Amsterdam, Netherlands. PA5154. 6. Iglarz M, et al. *J Cardiovasc Pharmacol.* 2015;66:457–67. 7. Sidharta PN, et al. *J Clin Pharmacol.* 2013;53:1131–8. 8. Weathers SP, et al. *Neurooncol Adv.* 2021;3:vdab141.

This material is distributed for scientific purposes on J&J Medical Connect, and is not for promotional use